2018 MEETING

ENCALS

European Network to Cure ALS

BOOK OF ABSTRACTS

20–22 JUNE 2018
OXFORD

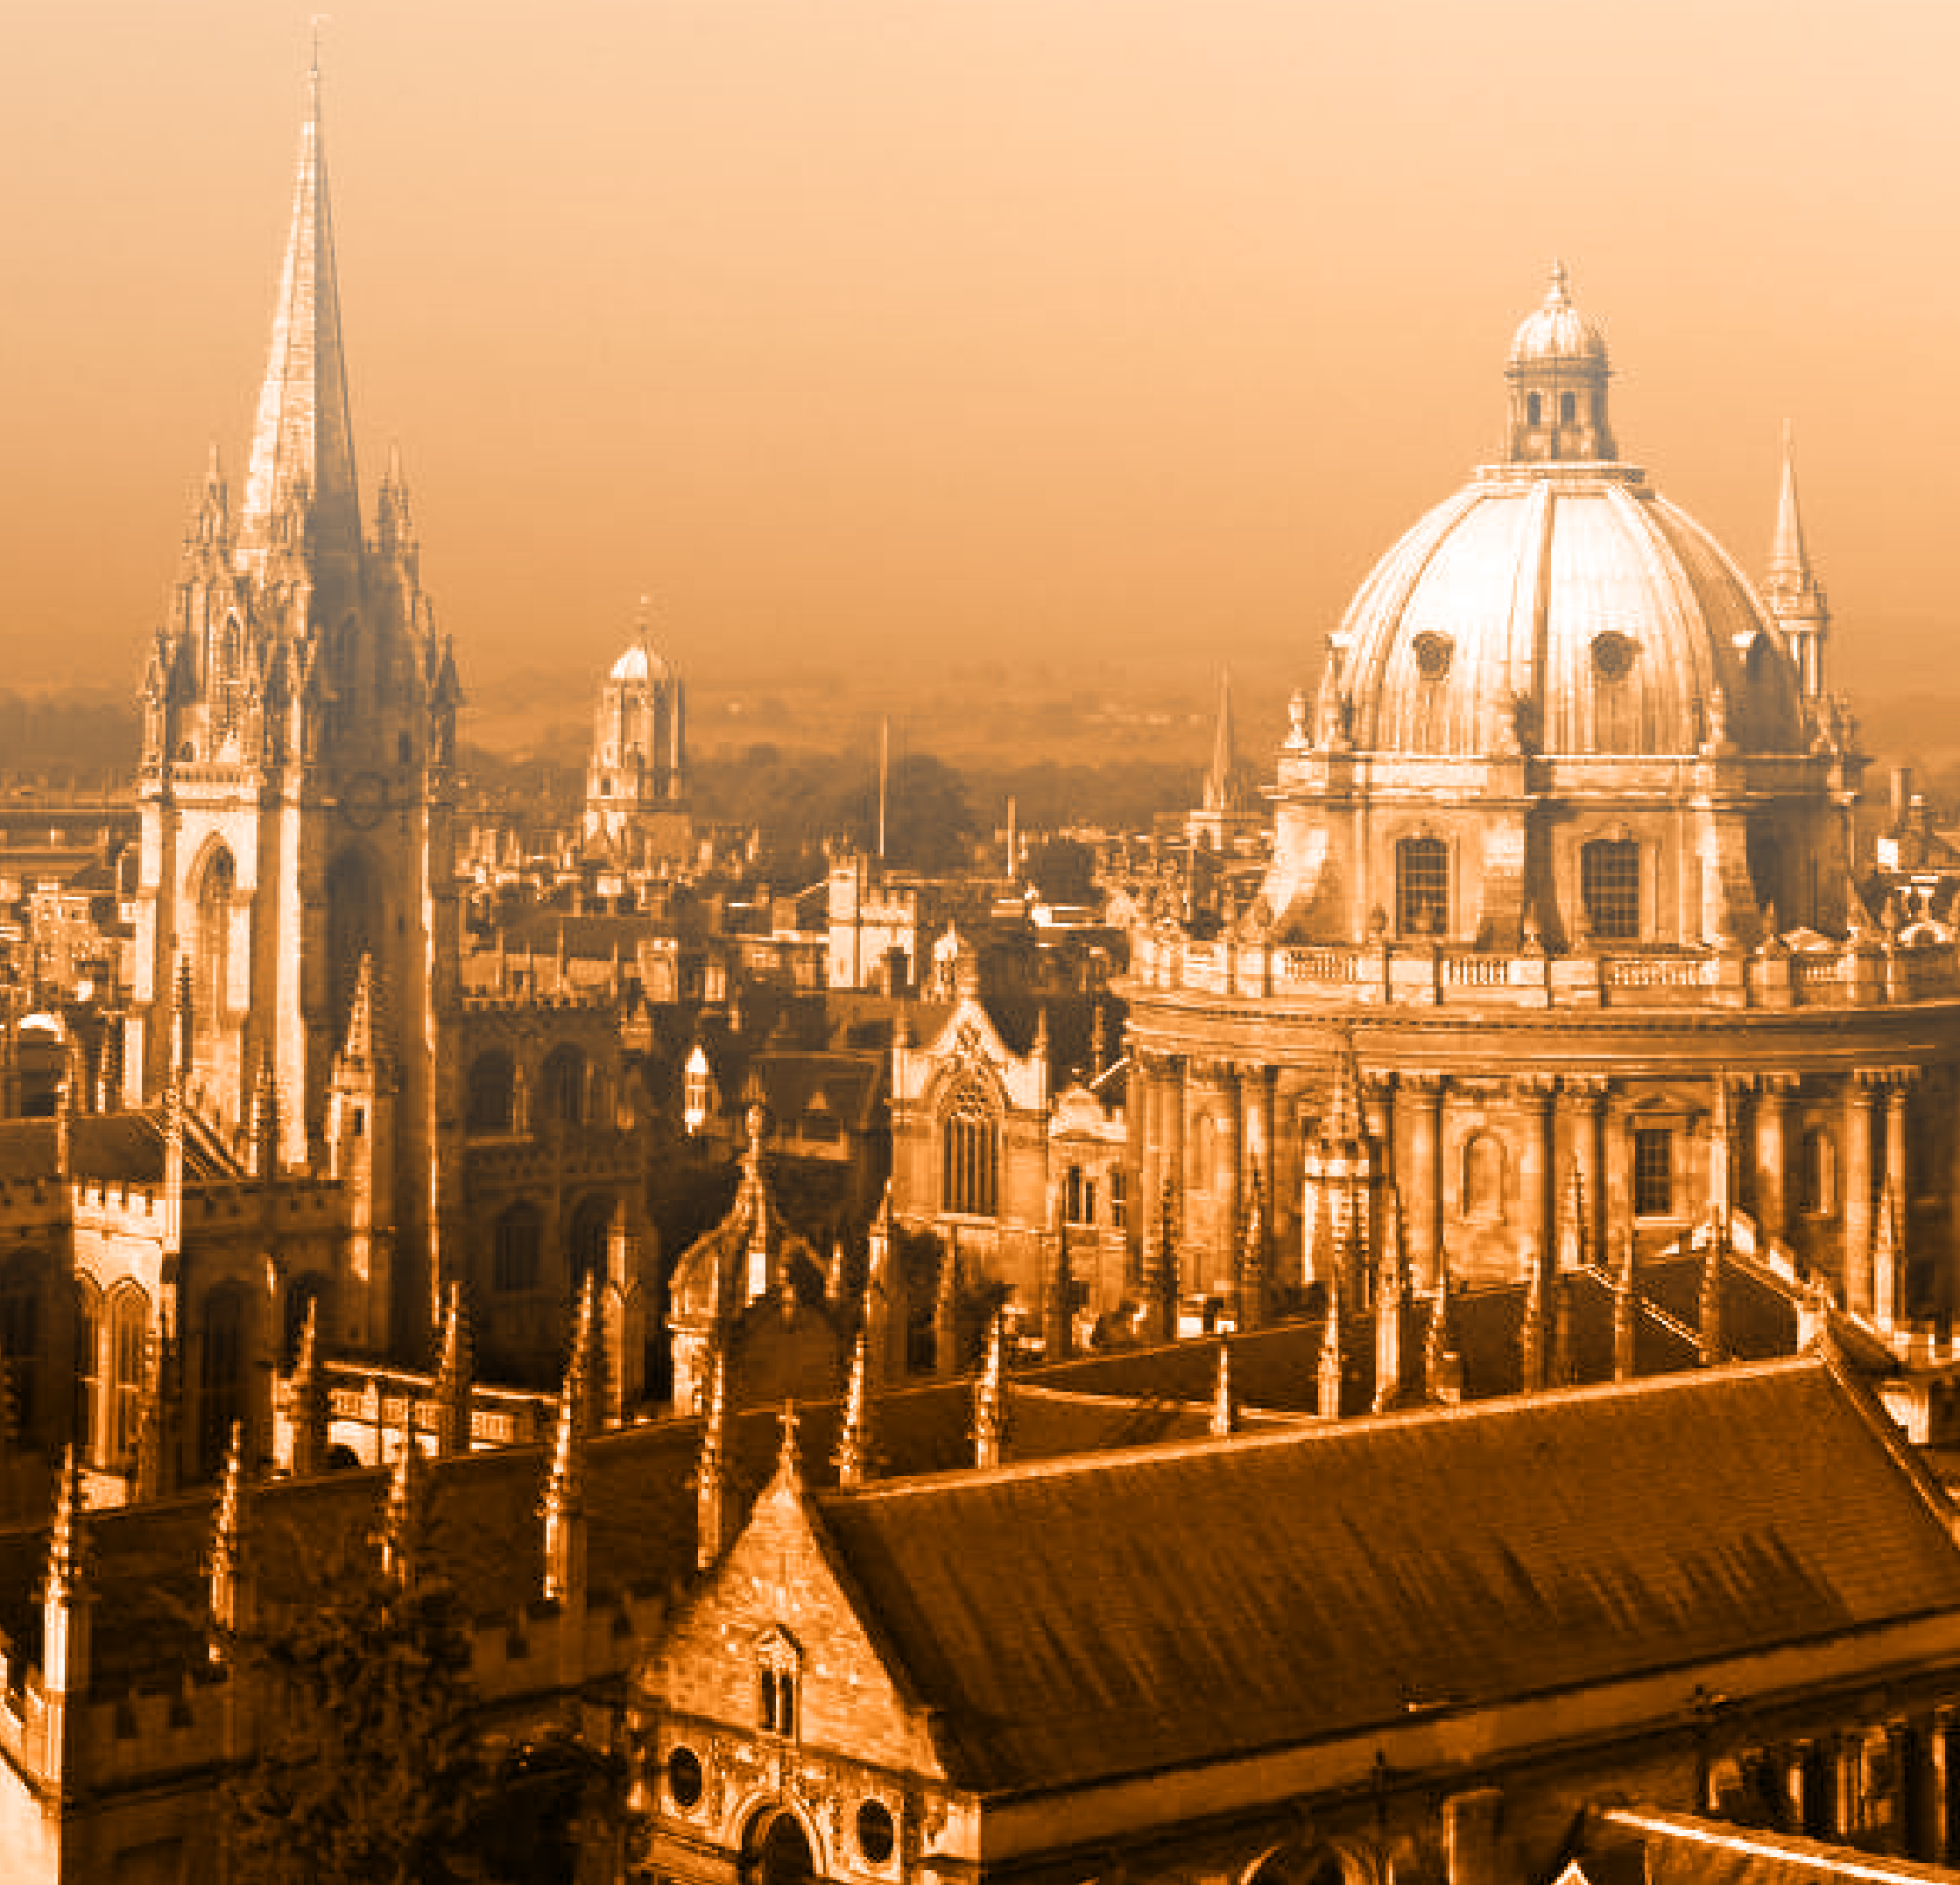

ENCALS Meeting 2018

C35 Initiation of masitinib at a less severe stage of disease produces greater treatment-effect: Subgroup analyses from masitinib study AB10015

Jesus S. Mora (1)*, Angela Genge (2), (On behalf of the AB10015 Study Group), Colin D. Mansfield (3), Olivier Hermine (3,4)

Masitinib (MAS) has previously reported positive phase 3 findings in ALS. Here we present post-hoc analyses that explored whether patient (pt) susceptibility to MAS is influenced by baseline disease severity, as measured by the individual component scores of ALSFRS-R. Study AB10015 used a prospectively stratified design based on ALSFRS-R progression rate calculated from disease-onset to baseline (ΔFS). A dichotomizing cutoff at 1.1 points/month distinguished between Normal Progressor (NP, ΔFS<1.1) and Fast Progressor (FP, ΔFS≥1.1) pts. The assumption here is that heterogeneity in ALS disease aggressiveness reflects differing disease mechanisms, leading to divergent treatment susceptibility. This approach therefore defines a more homogeneous primary efficacy population (i.e. NP pts receiving oral MAS 4.5 mg/kg/day), while concurrently permitting evaluation of the more heterogeneous population (i.e. 'NP plus FP' pts receiving MAS 4.5 mg/kg/day). The primary outcome endpoint, decline in ALSFRS-R from baseline to week-48 (ΔALSFRS-R), showed benefit for MAS (n=99) over placebo (PBO) (n=102) with a between-group least-squares means difference (ΔLSM) of 3.39 (-9.24 vs -12.63), corresponding to a significant and clinically meaningful slowing in functional decline of 27% (P=0.016). Conversely, significance on ΔALSFRS-R was not reached for the 'NP plus FP' cohort (secondary analysis), with a ΔLSM of 2.09 in favor of MAS (P=0.12). Post-hoc analysis showed that initiation of MAS treatment at a less severe stage of disease produced greater treatment-effect for both ΔFS-stratified cohorts in terms of ΔALSFRS-R (as well as secondary endpoints of FVC, ALSAQ-40, and survival-to-event analysis). Notably, this minor adjustment in pt selection criteria revealed a significant benefit for MAS in the 'NP plus FP' cohort. For pts with a baseline score of ≥1 on each ALSFRS-R item, ΔALSFRS-R was -9.8 for MAS (n=92) vs -13.1 for PBO (n=105); a ΔLSM of 3.3 and 25% slower rate of decline (P=0.0266). For pts with ≥2 on each ALSFRS-R item, ΔALSFRS-R was -6.7 for MAS (n=48) vs -11.5 for PBO (n=57); a ΔLSM of 4.8 and 42% slower rate of decline (P=0.0152). Finally, for pts with ≥3 on each ALSFRS-R item, ΔALSFRS-R was -4.3 for MAS (n=20) vs -15.1 for PBO (n=27); a ΔLSM of 10.8 and 72% slower rate of decline (P=0.0064). Results indicate greater benefit is possible when initiating MAS at a less severe stage of disease, with a significant treatment-effect seen regardless of post-onset ΔFS.

1) ALS Unit, Hospital San Rafael, Madrid, Spain.
2) ALS Unit, Montreal Neurological Institute and Hospital, Montreal, Canada.
3) AB Science, Paris, France.
4) Department of Hematology, Necker Hospital, University of Paris Descartes, France.

101

INITIATION OF MASITINIB AT A LESS SEVERE STAGE OF DISEASE PRODUCES GREATER TREATMENT-EFFECT: SUBGROUP ANALYSES FROM STUDY AB10015

Jesus S. Mora*[1], Olivier Hermine[2,3], Colin D. Mansfield[3], Angela Genge[4] (on behalf of the AB10015 Study Group)

*Corresponding author: sanrafael.neurologia@hsjd.es

[1]ALS Unit, Hospital San Rafael, Madrid, Spain. [2]Department of Hematology, Necker Hospital, University of Paris Descartes, France.
[3]AB Science, Paris, France. [4]ALS Clinic, Montreal Neurological Institute and Hospital, Montreal, Canada.

SUMMARY OF MASITINIB STUDY AB10015 DESIGN AND RESULTS

STUDY AB10015 WAS THE FIRST SUCCESSFULLY COMPLETED PLACEBO CONTROLLED PHASE 3 TRIAL OF MASITINIB IN ALS

Masitinib is an oral tyrosine kinase inhibitor that exerts a protective effect on both the CNS and PNS [Trais 2017; Trias 2016; see also ENCALS Poster C33].

AB10015 STUDY DESIGN

Prospectively stratified design based on ALSFRS-R progression (ΔFS)

- Defines homogenous target population for primary analysis (patient enrichment criteria).
- This approach and defined cut-off supported by the literature [Gordon, 2006; Kimura, 2006; Kollewe, 2008; Labra, 2016].
- Study enrichment leads to lower sample size / increased power for the trial.
- Concurrently permits evaluation (secondary) of the broader population.
- Rationale: heterogeneity in ALS disease aggressiveness reflects differing disease mechanisms, with possible divergent treatment-effect.

- Double blind, controlled, randomized 1:1:1, oral, 48 week treatment
 - Masitinib 4.5 mg/kg/day + riluzole (high-dose cohort) vs. Placebo + riluzole
 - Masitinib 3.0 mg/kg/day + riluzole (low-dose cohort) vs, Placebo + riluzole
- Main inclusion criteria:
 - Lab-supported probable, probable, or definite ALS; sporadic or familial
 - Disease duration <36 months, FVC ≥60%, stable riluzole dosing
- Two distinct patient populations
 - ‘Normal Progressors’: progression rate <1.1 points/month
 - ‘Fast Progressors’: progression rate ≥1.1 points/month
- Four distinct efficacy datasets
 - Primary efficacy population predefined as the ‘Normal Progressor’ cohort receiving 4.5 mg (high-dose) masitinib
- Efficacy analysis endpoints:
 - Primary – Decline in ALSFRS-R from baseline to week-48
 - Secondary included – Survival-to-event analysis (a surrogate measure for overall survival) defined as the time interval (months) for a ALSFRS-R deterioration of 9 points from baseline or death.

KEY RESULTS FROM HIGH-DOSE COHORT

Masitinib (4.5 mg/kg/day) demonstrated a significant benefit in ALS patients with a ΔFS of <1.1 points/month (primary analysis population)

	n	LSM	ΔLSM [95%CI]	Effect	*P* value
ALSFRS-R (Primary analysis)					
PBO	102	-12·6	3·4 [0·7 ; 6·1]	27%	0·016
M4·5	99	-9·2			
SURVIVAL-TO-EVENT (Secondary analysis)			Median[95%CI]	Effect	*P* value
PBO	113		16 months [11;19]	25%	0·016
M4·5	105		20 months [14;30]		

However, significance was not reached in the more heterogeneous, population (secondary analysis) of the ‘Normal and Fast Progressor’ masitinib (4.5 mg/kg/day) cohort

	n	LSM	ΔLSM [95%CI]	Effect	*P* value
ALSFRS-R (Primary analysis)					
PBO	119	-13.0	2.09 [-0.5 ; 4.7]	16%	0·12
M4·5	120	-10.9			
SURVIVAL-TO-EVENT (Secondary analysis)			Median[95%CI]	Effect	*P* value
PBO	132		14 months [11;17]	21%	0·044
M4·5	128		17 months [14;22]		

SUBGROUP ANALYSIS EXPLORING EFFECT OF BASELINE DISEASE SEVERITY

EXPLORATORY ANALYSES SHOWED GREATER BENEFIT IS POSSIBLE WHEN INITIATING MASITNIIB AT A LESS SEVERE STAGE OF DISEASE

- Post-hoc analyses explored whether treatment-effect was influenced by baseline disease severity, as measured by ALSFRS-R individual item scores.
 - Exclusion of patients with zero-point ALSFRS-R items scores (i.e. very severely affected) is a credible clinical scenario in which treatment is initiated prior to severe symptom onset and better matches clinical practice.
 - Logical trial design feature considering that any ALSFRS-R component scoring zero at baseline cannot register any slowing of disease (i.e. insensitive to masitinib treatment-effect).
- Results showed that initiation of masitinib at a less severe stage of disease produced greater treatment-effect for both ΔFS-stratified high-dose cohorts.
 - Notably, this minor adjustment in patient selection criteria revealed a significant benefit for masitinib over placebo in the broader (‘Normal and Fast Progressor’) masitinib 4.5 mg/kg/day cohort.
 - Fast Progressors well-represented and balanced in the ALSFRS-R item ≥1 cohort (M4.5 = 12 pts vs. PBO = 11 pts).
 - Positive treatment-effect demonstrated for decline in ALSFRS-R (primary endpoint) and several secondary endpoints such as survival-to-event analysis, FVC, and ALSAQ-40.
- Sensitivity analyses based on full analysis dataset (non-LOCF) imputation methods were also positive with similar results.

- Results of subgroup analysis exploring effect of baseline disease severity, as measured by the individual component scores of ALSFRS-R, where a higher threshold indicates less severe disease.
 - ALSFRS-R analysis in ‘Normal & Fast Progressor’ (4.5 mg/kg/d) cohort

ALSFRS-R ANALYSIS (‘Normal and Fast Progressor’ masitinib 4.5 cohort)

		n	LSM	ΔLSM [95%CI]	ΔEffect	*P* value
Severe, Moderate, Mild	ALSFRS-R item ≥1					
	PBO	105	-13.1	3.27 [0.4 ; 6.1]	25%	0.0266
	M4.5	92	-9.83			
Moderate and Mild	ALSFRS-R item ≥2					
	PBO	57	-11.51	4.81 [0.9 ; 8.7]	42%	0.0152
	M4.5	48	-6.70			
Mild	ALSFRS-R item ≥3					
	PBO	27	-15.1	10.8 [3.2 ; 18.4]	72%	0.0064
	M4.5	20	-4.3			

 - Survival-to-event analysis in ‘Normal & Fast Progressor’ (4.5 mg/kg/d) cohort

SURVIVAL-TO-EVENT ANALYSIS (‘Normal and Fast Progressor’ masitinib 4.5 cohort)

	n	Median[95%CI]	ΔMedian	*P*-value
ALSFRS-R item ≥1				
PBO	115	16 [11; 19]	4 months	0.022
M4.5	96	20 [14; 30]		
ALSFRS-R item ≥2				
PBO	63	17 [11; 33]	13 months	0.1502
M4.5	50	30 [15; NR]		
ALSFRS-R item ≥3				
PBO	28	11 [8.3; 19]	19 months	0.0071
M4.5	22	30 [22; NR]		

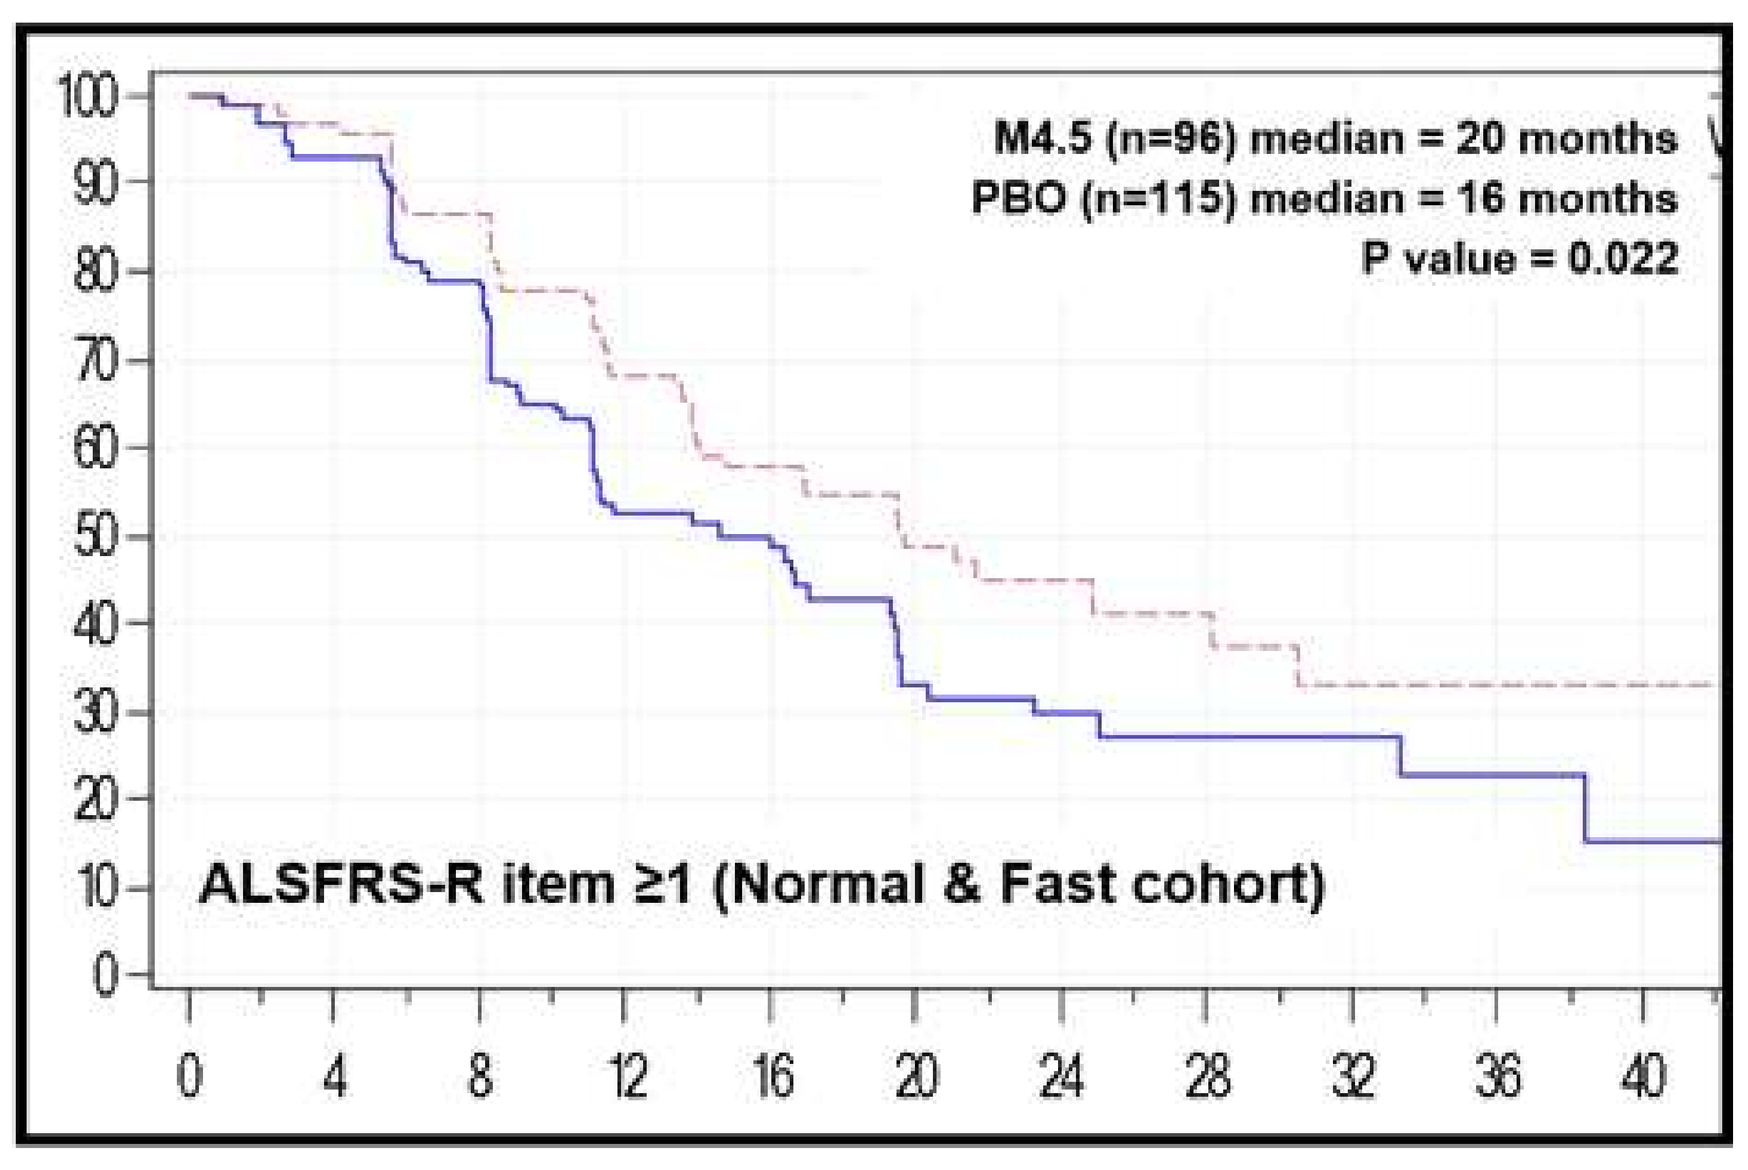
M4.5 (n=96) median = 20 months
PBO (n=115) median = 16 months
P value = 0.022
ALSFRS-R item ≥1 (Normal & Fast cohort)
0 10 20 30 40 50 60 70 80 90 100
0 4 8 12 16 20 24 28 32 36 40

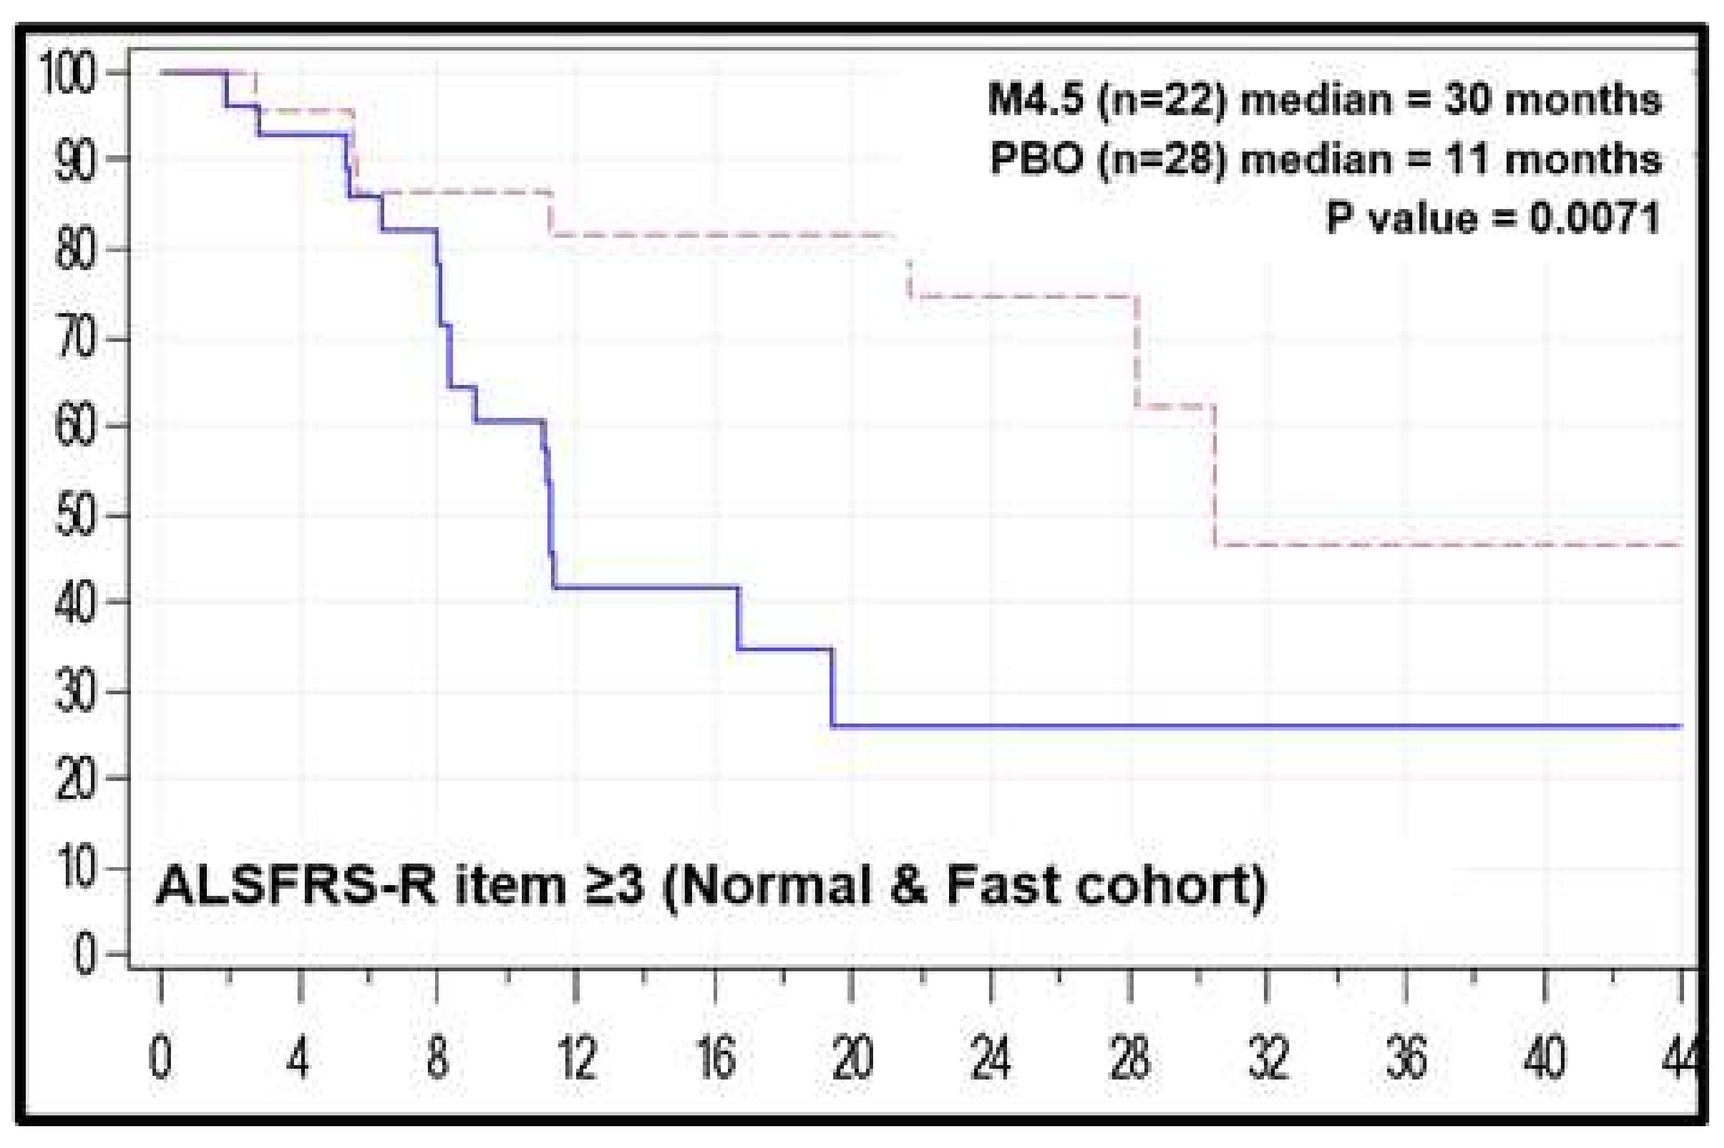
M4.5 (n=22) median = 30 months
PBO (n=28) median = 11 months
P value = 0.0071
ALSFRS-R item ≥3 (Normal & Fast cohort)
0 10 20 30 40 50 60 70 80 90 100
0 4 8 12 16 20 24 28 32 36 40 44

These findings have revealed a relevant design consideration for future clinical studies of masitinib in ALS.

Together with patient selection based on ΔFS, such clinical trial enrichment strategies are important for efficient study design in ALS.

References: Trias (2016) J Neuroinflammation; 13(1):177. Trias (2017) JCI Insight; 2(20):e95934. Gordon (2006) Neurology; 66:1117–9. Kimura (2006) Neurology; 66:265–7. Kollewe (2008) J Neurol Sci; 275:69–73. Labra (2016) J Neurol Neurosurg Psychiatry; Jun;87(6):628-32.

ENCALS Annual Meeting 2018 Oxford, UK (20-22 June 2018)