EUROPEAN
HEMATOLOGY
ASSOCIATION

MADRID

22ND CONGRESS

JUNE 22 – 25 | 2017

European Hematology Association

FINAL PROGRAM

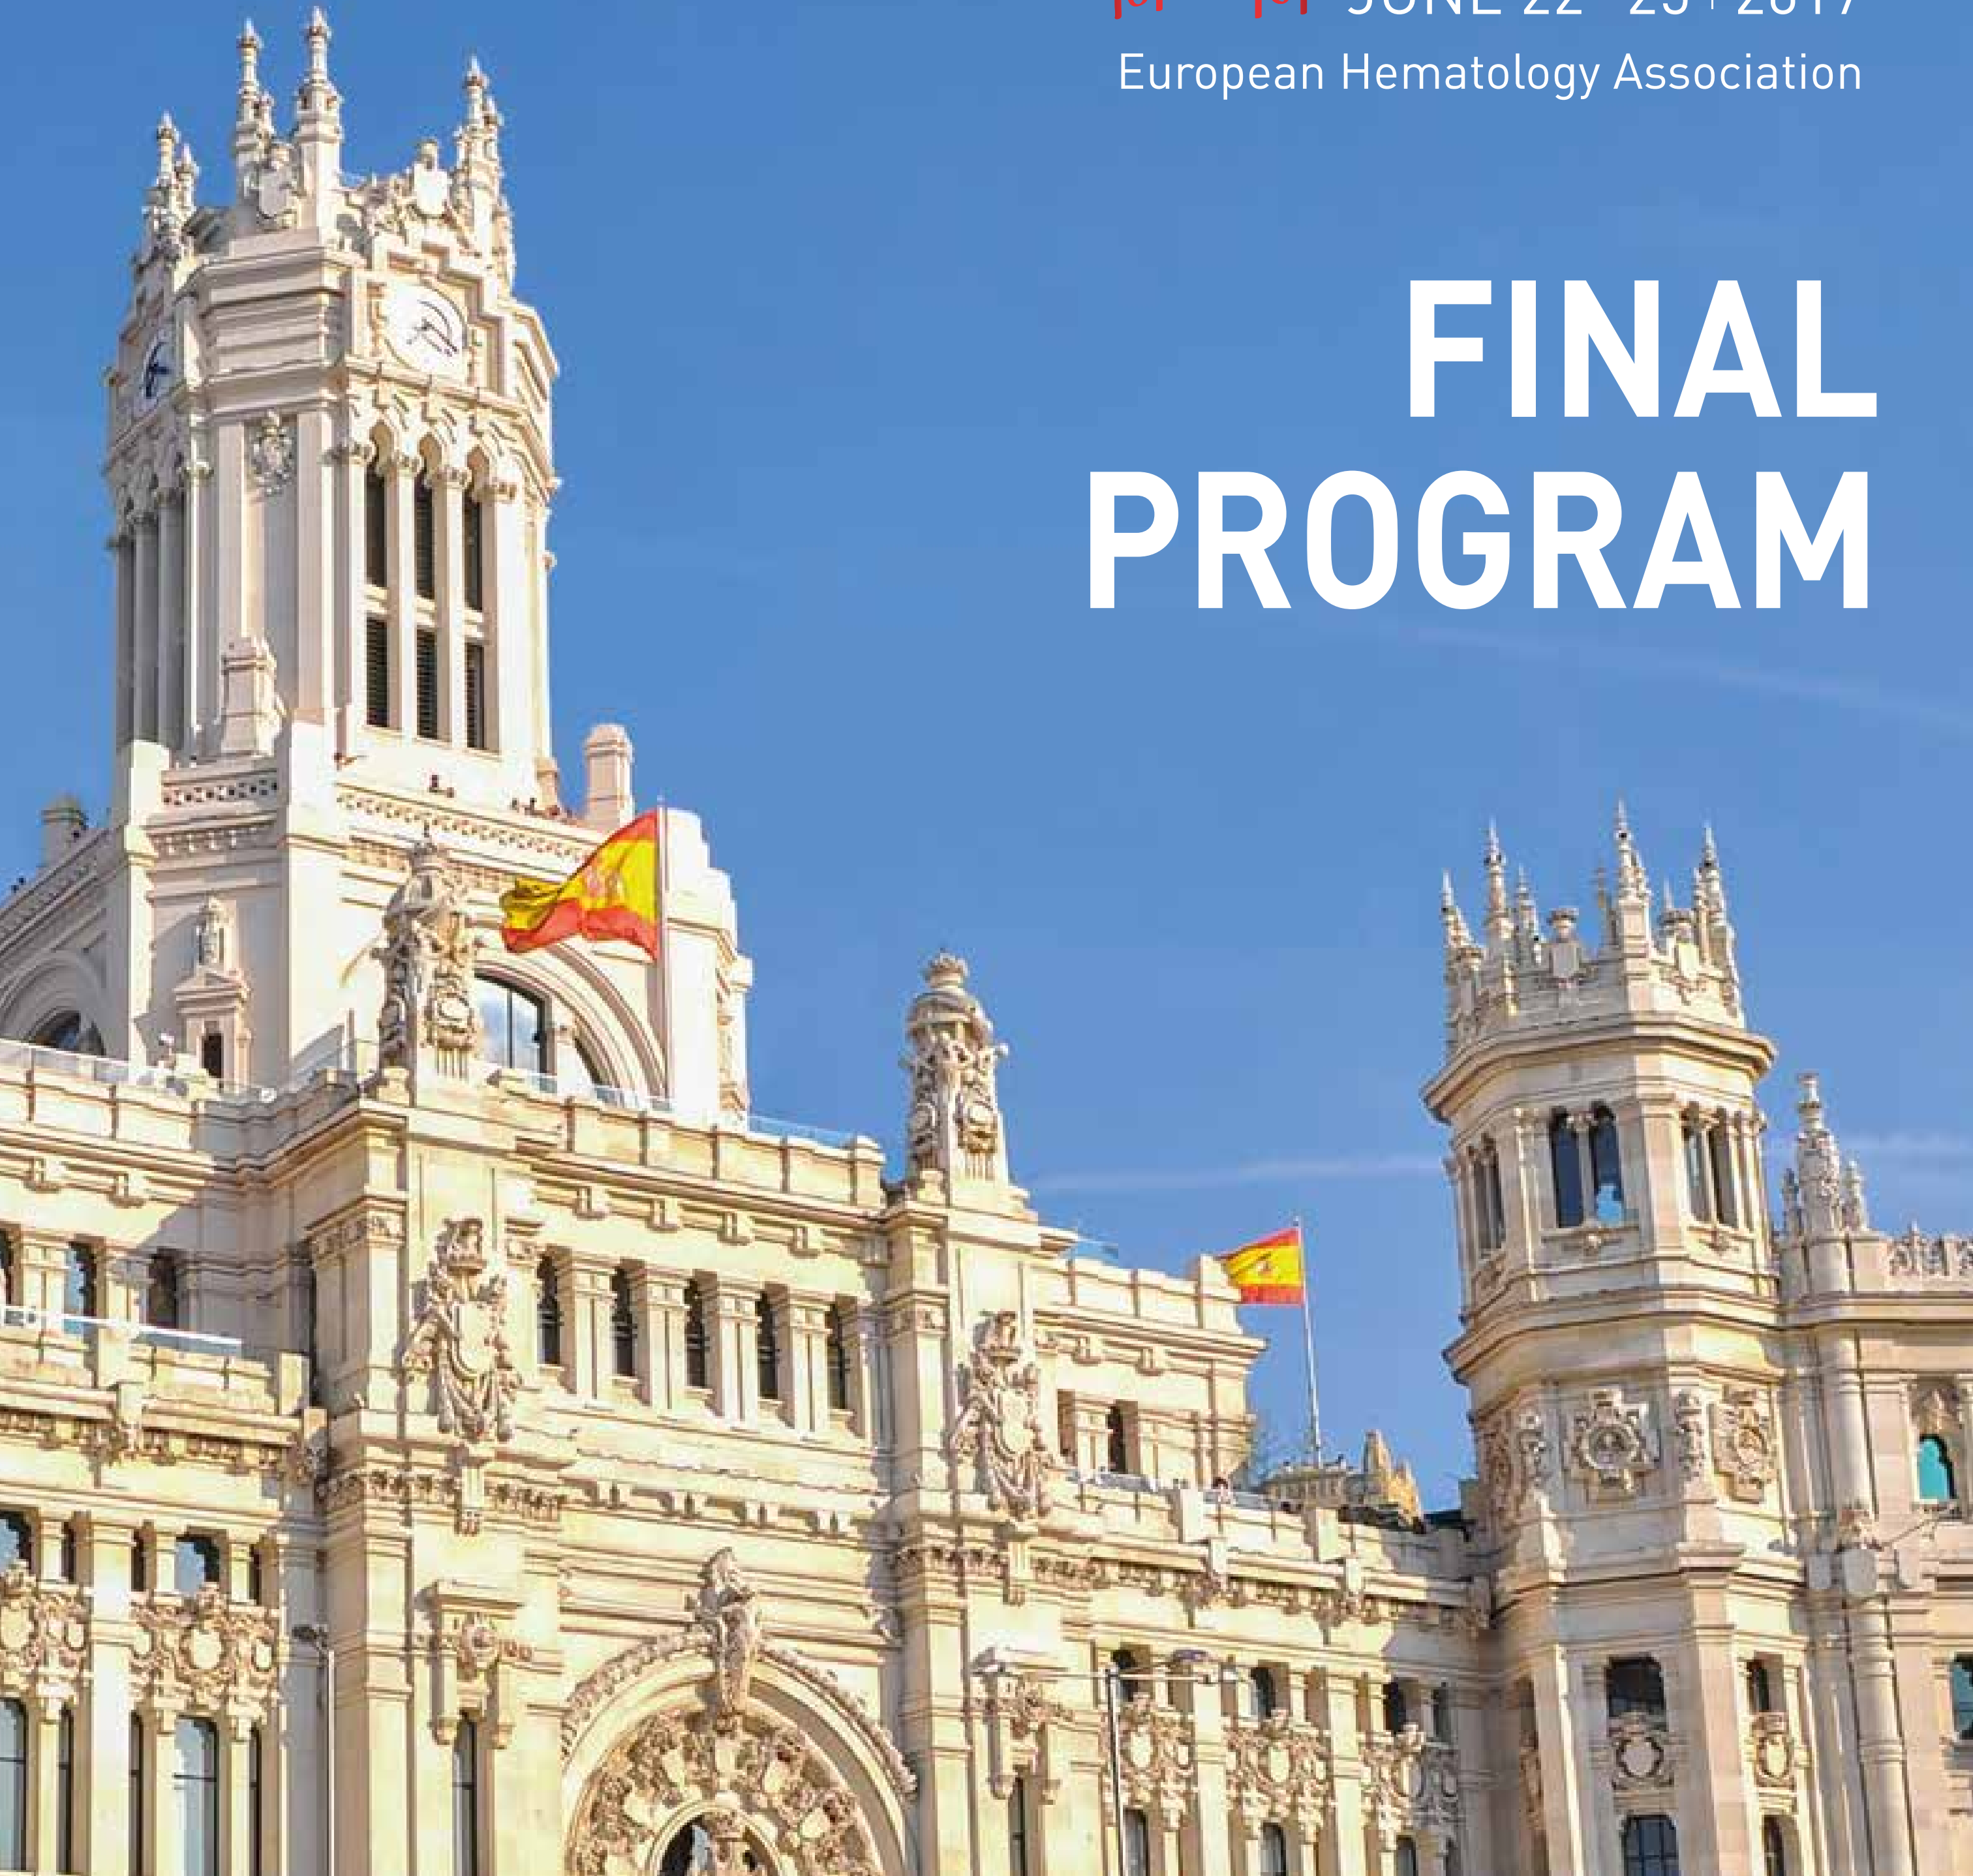

EHA 2017

MASITINIB FOR TREATMENT OF SEVERELY SYMPTOMATIC INDOLENT SYSTEMIC MASTOCYTOSIS: ADDITIONAL EFFICACY ANALYSES FROM THE RANDOMIZED, PLACEBO-CONTROLLED, PHASE 3 STUDY

Background: Masitinib, a selective oral tyrosine kinase inhibitor targeting wild-type KIT, LYN and FYN, was the first drug to demonstrate efficacy in a phase 3 setting (study AB06006) for treatment of patients with severe indolent systemic mastocytosis (ISM) who are unresponsive to existing, optimal symptomatic treatments. In *The Lancet* (online Jan 6, 2017), Lortholary and colleagues reported a significant and clinically meaningful treatment benefit for masitinib (6 mg/kg/day over 24-weeks) versus placebo, with primary analysis based on cumulative response (≥75% improvement from baseline, timeframe weeks 8–24, comprising 5 visits at 4-week intervals) in at least one of four severe baseline symptoms (pruritus, flushes, depression, or fatigue) using repeated measures methodology for rare diseases (i.e. a longitudinal analysis with respect to symptoms as opposed to patient response rate at a single point in time). Eligible patients were aged 18–75 years and had ISM according to inclusion criteria that were slightly broader than the WHO classification.

Aims: To aide interpretation of this study's prospectively declared primary endpoint via comparison with additional efficacy analyses based on a cohort restricted to the WHO classification of ISM and more conventional patient-centric response endpoints.

Methods: Randomized, placebo-controlled, phase 3 study that included 135 severely symptomatic ISM patients, including the subvariant smoldering systemic mastocytosis (71 masitinib, 64 placebo), 80% of whom satisfied the WHO classification.

Results: Masitinib showed a significant improvement over placebo according to its pre-specified primary endpoint (mITT population), with a cumulative response of 18.7% versus 7.4%, respectively, odds ratio (OR) of 3.6 [95%CI 1.2-10.8], P=0.008 (with re-randomization). This outcome was confirmed in the WHO patient subgroup: 17.8% versus 8.0%, respectively, OR=3.25 [0.97-10.88], P=0.0317. Computing the primary analysis (mITT) according to cumulative response per patient (GEE model) was also positive: 26.7% versus 12.8%, respectively, OR=2.48 [1.16-5.31], P=0.0212; as was analysis according to individual patient response (Pearson chi-square): 40.3% versus 24.2%, respectively, P=0.0062. Response (per patient) on all severe baseline symptoms for at least one visit was: 16.4% versus 1.6%, respectively, P=0.0062. Finally, analysis of sustained response in all severe baseline symptoms over multiple visits was highly discriminatory between treatment-arms: for patients with 3 severe baseline symptoms masitinib generated a 12.5% response rate (≥75% improvement in each symptom) for 3 out of 5 visits, versus no response for placebo; and for patients with 2 severe baseline symptoms masitinib generated a response rate of 21.1%, 15.8% and 10.5% over at least 1, 2, and 3 visits, respectively, versus no response for placebo.

Summary/Conclusion: These post-hoc analyses confirm the clinical relevance, durability, and generalizability of the positive primary endpoint from study AB06006. Findings therefore support the conclusion that masitinib generates a significant therapeutic benefit in patients with severely symptomatic ISM who were unresponsive to optimal symptomatic treatments.

Email address: ohermine@gmail.com

Keywords: Mast cell disease, Mastocytosis, Phase III, Treatment, Masitinib

Masitinib for Treatment of Severely Symptomatic Indolent Systemic Mastocytosis: Additional Efficacy Analyses from the Randomized, Placebo-Controlled, Phase 3 Study

O Hermine[1, 2, 3], C Paul[4], E Jassem[5], M Niedoszytko[5], S Barete[6], S Verstovsek[7], C Grattan[8], D Canioni[9], S Georgin-Lavialle[10], L Frenzel[1], JP Kinet[11], C Mansfield[3], A Moussy[3], P Dubreuil [3,12,13,14], O Lortholary[15, 16]

Corresponding author: Olivier Hermine MD, PhD. E-mail: ohermine@gmail.com

[1]University Paris Descartes, Institut Imagine INSERM U1163, Paris, France; [2]CNRS ERL8654, Centre de Référence des Mastocytoses (CEREMAST), Paris, France; [3]AB Science, Paris, France; [4]Paul Sabatier University, Hôpital Larrey, Toulouse, France; [5]Medical University of Gdansk, Gdansk, Poland; [6]Hôpital Tenon, Paris, France; [7]Hanns A. Pielenz Clinical Research Center for Myeloproliferative Neoplasms, Houston, United States; [8]Norfolk & Norwich University Hospital, Norwich, United Kingdom; [9]Université Paris Descartes, Paris Sorbonne Cité, APHP Necker-Enfants Malades; [10]Hôpital Tenon, Université Pierre et Marie Curie, Paris, France; [11]Harvard Medical School & Beth Israel Deaconess Medical Center, Boston, United States; [12]Inserm U1068, CRCM; [13]CNRS, UMR7258, Institut Paoli-Calmettes, Aix-Marseille Université, Marseille; [14] CEREMAST, Paris, France; [15]Hôpital Necker-Enfants-Malades, Université Paris Descartes, Sorbonne Paris Cité, [16] CEREMAST, Paris, France.

Introduction

➢ ***Indolent systemic mastocytosis (SM), including smoldering SM, is a rare disease with a wide variety of clinical manifestations***

- About 33% of indolent SM patients will experience severely debilitating symptoms (handicaps) that are unresponsive to optimal symptomatic treatment.
- There is a high unmet medical need for these severely symptomatic, treatment-refractory patients.

➢ ***Masitinib is an oral tyrosine kinase inhibitor that inhibits mast cell activation, degranulation and migration***

- Masitinib selectively targets kinases that play critical roles in mast cell (MC) function.

INHIBITORY EFFECTS OF MASITINIB AND KINOME INTERACTION MAPS

Target	IC_{50} [nM]	Kd [μM]
KIT wild-type (WT)	200	0.008
FYN	240	0.14
LYN	225	0.061
KIT JM mutation (exon 11)	5	
KIT-D86V (exon 17)	5,000	

TREESPOT KINOME INTERACTION MAPS

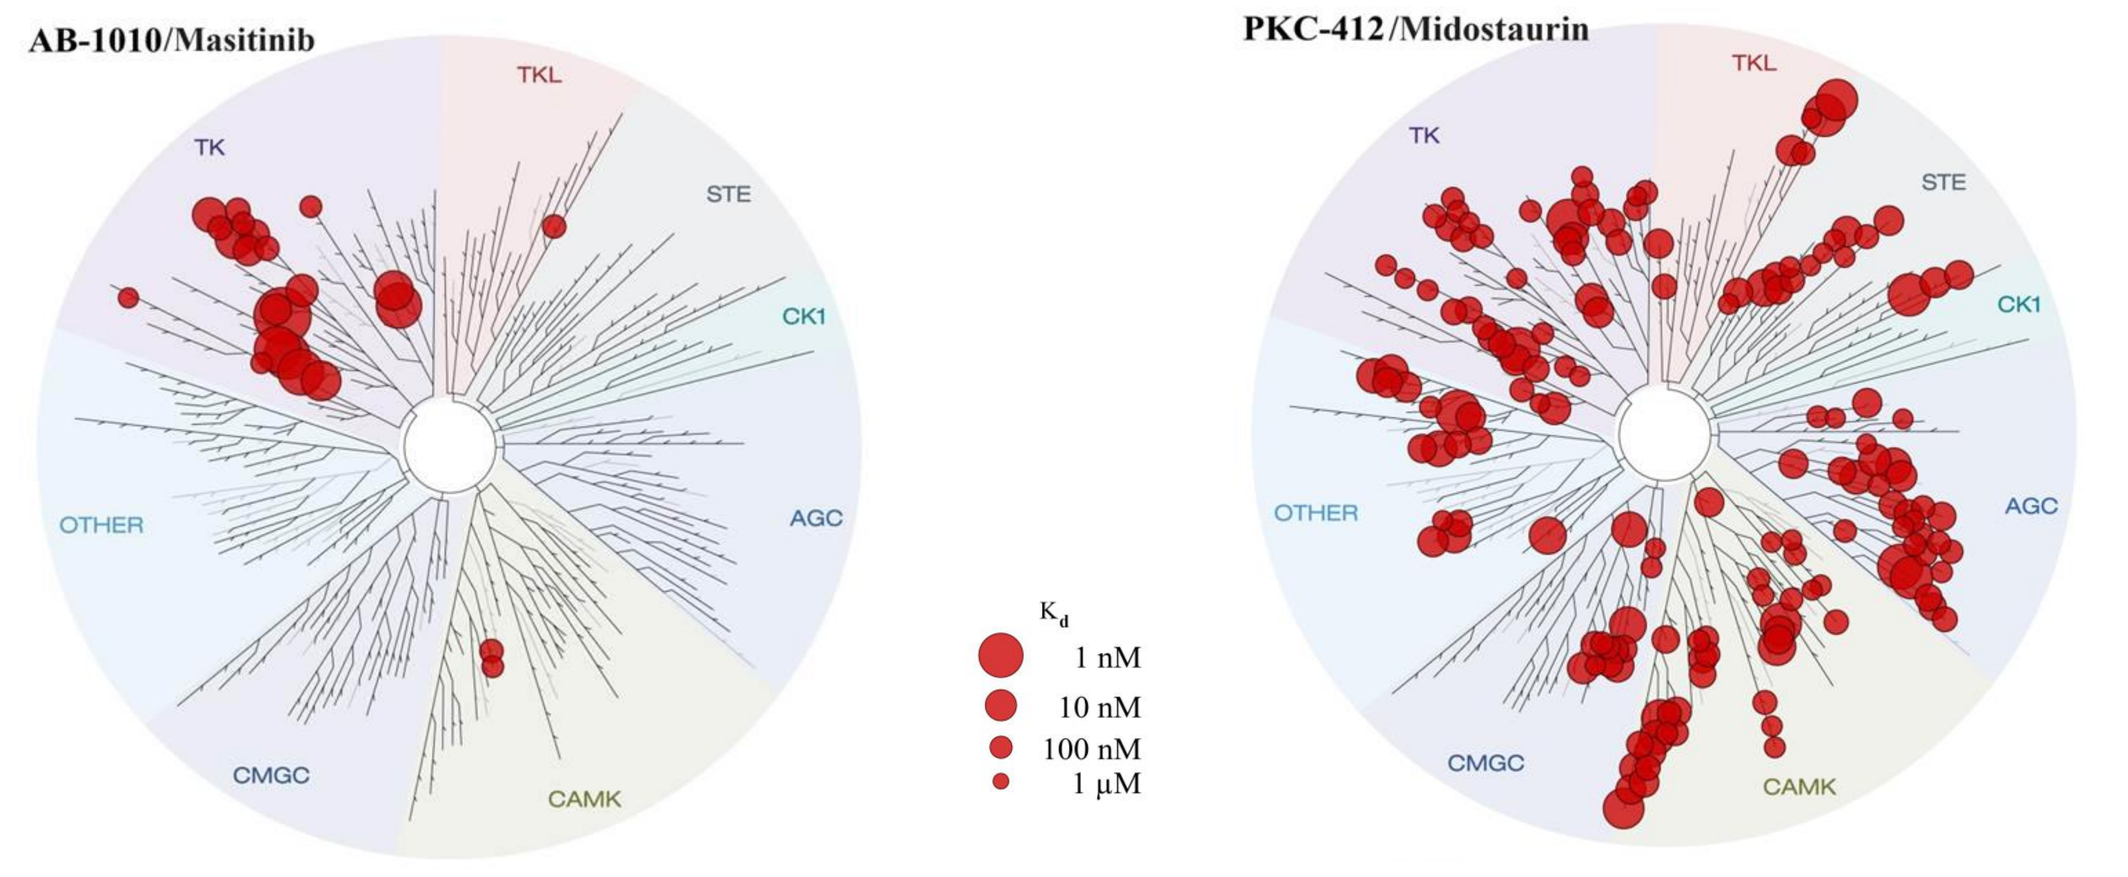
AB-1010/Masitinib
PKC-412/Midostaurin
TKL
TK
STE
CK1
AGC
OTHER
CMGC
CAMK
K_d
1 nM
10 nM
100 nM
1 μM

- Masitinib's action relies on targeting normal (WT) MCs and/or reducing activation of KIT-D816V MCs; the latter via modulating MC degranulation in a KIT-D816V-independent manner,
- Masitinib's high kinase selectivity limits the risk of off-target toxicity.

Study Design & Results in Context

➢ ***Masitinib study AB06006 is the first phase 3, prospective, randomized controlled trial for a treatment of indolent SM in patients unresponsive to existing, optimal symptomatic treatment***

- Eligible patients (pts) had indolent SM according to slightly broader criteria than the WHO classification, and at least one severe baseline symptom (handicap) from among: pruritus score ≥9, flushes/week ≥8, Hamilton depression rating ≥19, or Fatigue Impact Scale ≥75.
- A total of 135 pts with severely symptomatic indolent SM were enrolled (71 masitinib, 64 placebo), 80% of whom satisfied the WHO classification (i.e. 27 pts did not fulfil the WHO classification).
- Treatment-effect tested using repeated measures methodology for rare diseases via the generalized estimating equation (GEE) model.
 - Primary endpoint was cumulative response in at least one severe symptom with response defined as a ≥75% improvement in handicap.
 - Cumulative response was defined as the number of actual responses between weeks 8 and 24, divided by the total number of possible responses over that period; i.e. longitudinal analysis with respect to symptoms as opposed to pt response rate at a single time-point.

➢ ***Positive findings have been reported in The Lancet showing a significant and clinically meaningful treatment benefit, with demonstrated possibility of effective, safe long-term management.[1]***

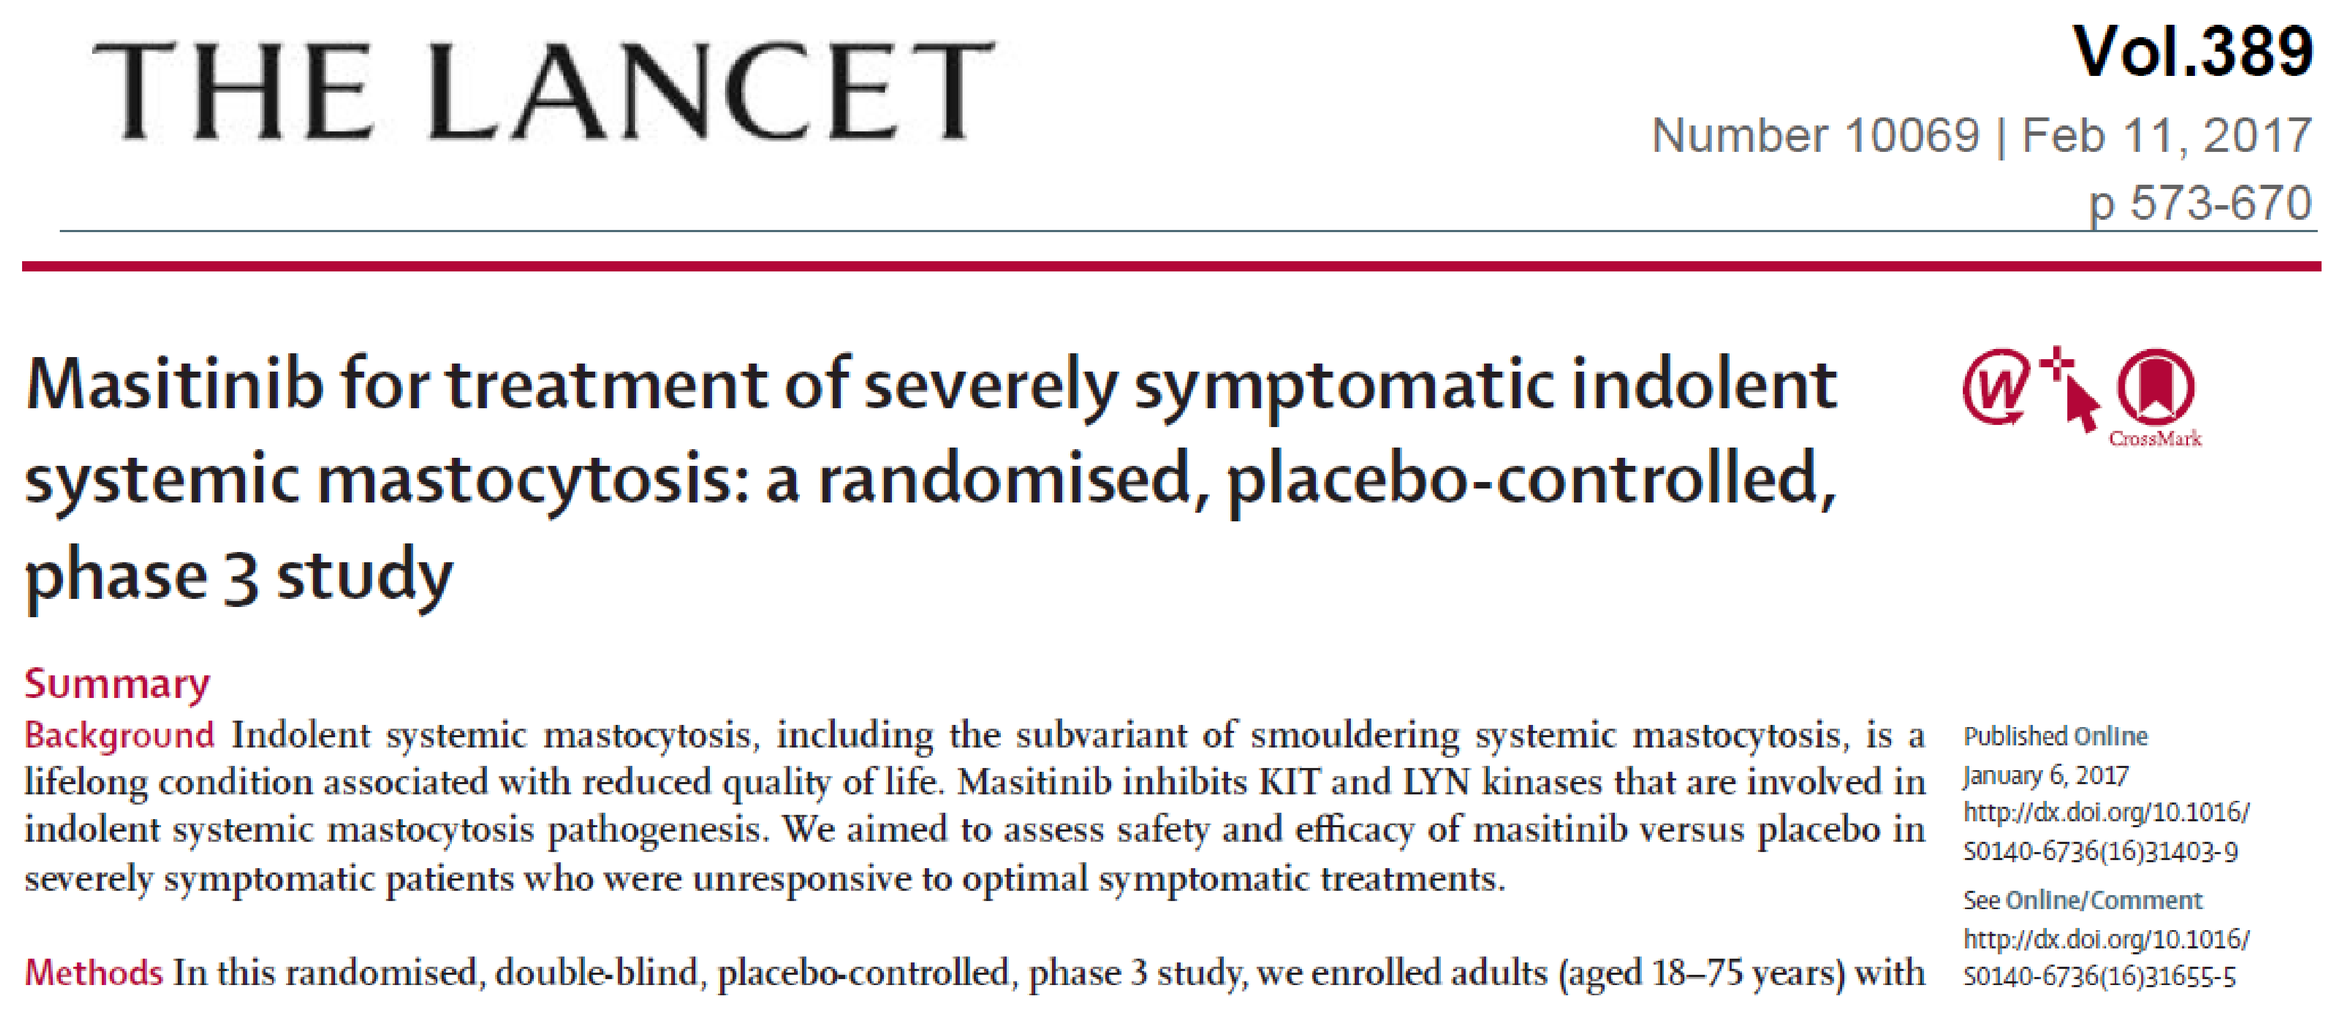
THE LANCET

Vol.389
Number 10069 | Feb 11, 2017
p 573-670

Masitinib for treatment of severely symptomatic indolent systemic mastocytosis: a randomised, placebo-controlled, phase 3 study

Summary

Background Indolent systemic mastocytosis, including the subvariant of smouldering systemic mastocytosis, is a lifelong condition associated with reduced quality of life. Masitinib inhibits KIT and LYN kinases that are involved in indolent systemic mastocytosis pathogenesis. We aimed to assess safety and efficacy of masitinib versus placebo in severely symptomatic patients who were unresponsive to optimal symptomatic treatments.

Methods In this randomised, double-blind, placebo-controlled, phase 3 study, we enrolled adults (aged 18–75 years) with

Published Online January 6, 2017 http://dx.doi.org/10.1016/S0140-6736(16)31403-9

See Online/Comment http://dx.doi.org/10.1016/S0140-6736(16)31655-5

➢ ***Masitinib showed significant improvement over placebo*** in its primary endpoint, 18.7% vs. 7.4%, respectively, (P=0.008).

- Success in the primary analysis was corroborated by secondary endpoints such as a significant change in tryptase level between treatment-arms (-18.0% vs. +2.2%, respectively, P<0.001).
- Data showed a positive benefit/risk ratio. Masitinib may therefore be an important new treatment option for these pts.

Supportive Analyses Confirm Results are Clinically Relevant

➢ ***Additional efficacy analyses based on a cohort restricted to the WHO classification of indolent SM and more conventional patient-oriented response endpoints confirm a consistent superiority of masitinib over placebo***

- Patient response based on the primary analysis but computed with respect to response per patient (as opposed to [pt * handicap * visit] or [pt * visit]) was consistently significant, corroborating the primary analysis based on the 4R75% response.
- Unlike masitinib, placebo was unable to generate a response for more than one visit.. Analysis of sustained response in all severe baseline symptoms over multiple visits was highly discriminatory between treatment-arms:
 - For pts with 3 severe baseline symptoms masitinib generated a 12.5% response rate (≥75% improvement in each symptom) for 3 out of 5 visits, vs. no response for placebo.
 - For pts with 2 severe baseline symptoms masitinib generated a response rate of 15.8% and 10.5% over at least 2 and 3 visits, respectively, vs. no response for placebo.

Analysis		Masitinib	Placebo	p-value
Primary (AB06006)	4R75% (GEE: Pt * handicap * visit)	18.7%	7.4%	0.0076
Primary (WHO)	4R75% (GEE: Pt * handicap * visit)	17.8%	8.0%	0.0317
Primary (per pt*visit)	4R75% (GEE: Pt * visit)	26.7%	12.8%	0.0212
Overall pt response	4R75% (Pearson Chi-Square: Pts)	40.3%	24.2%	0.0062
Overall pt response	Response on all baseline handicaps	16.4%	1.6%	0.0038
Pt with 2 handicaps	Response on each baseline handicap	21.0% (n=19)	0.0% (n=25)	N/A
Pt with 3 handicaps	Response on each baseline handicap	12.5% (n=16)	0.0% (n=18)	N/A
Pt with 4 handicaps	Response on each baseline handicap	16.7% (n=6)	0.0% (n=3)	N/A

Primary (AB06006) = prospectively declared primary endpoint. 4R75% = Cumulative 75% response rate on pruritus or flushes or depression or asthenia. GEE = generalized estimating equation model. mITT population: masitinib n = 67; placebo n = 62. Response defined as a ≥75% improvement in severe baseline handicap. N/A not applicable.

➢ ***These findings support the conclusion that masitinib generates a significant therapeutic benefit in patients with severely symptomatic indolent SM, who are unresponsive to optimal symptomatic treatments***

- Additional analyses aide interpretation of the study's prospectively declared primary endpoint using more conventional measures of response or WHO inclusion criterion.
- Data confirm the clinical relevance, durability, and generalizability of the positive primary endpoint from study AB06006.

References: [1] Lortholary et al. Lancet. 2017 Feb 11;389(10069):612-620.

22nd Congress of the European Hematology Association — June 22-25, 2017, Madrid, Spain