Masitinib significantly slows disease progression in post-paralysis SOD1^{G93A} rats and reduces inflammation in both CNS and PNS

Emiliano Trias[1], Sofía Ibarburu[1], Romina Barreto-Núñez[1], Joël Babdor[2], Thiago Maciel[2], Pablo Díaz-Amarilla[3], Patricia Cassina[4], Laura Martínez-Palma[4], Colin Mansfield[5], Alain Moussy[5], Ivan Moura[2], Joseph Beckman[6], Olivier Hermine[2,7], Luis Barbeito[1]

[1]Institut Pasteur de Montevideo, Uruguay; [2] Imagine Institute, Hôpital Necker, Paris, France;[3]Instituto de Investigaciones Biológicas Clemente Estable, Montevideo, Uruguay; [4]Departamento de Histología, Facultad de Medicina, UdelaR, Montevideo, Uruguay; [5]ABScience [6]Linus Pauling Institute, Department of Biochemistry and Biophysics, Oregon State University [7]Department of Hematology, Necker Hospital; INSERM UMR 1163, Laboratory of cellular and molecular mechanisms of hematological disorders and therapeutic implications; Paris Descartes – Sorbone Paris Cité University, Imagine Institute; CNRS ERL 8254; Laboratory of Excellence GR-Ex; Centre national de référence des mastocytoses (CEREMAST), Paris, France

Introduction

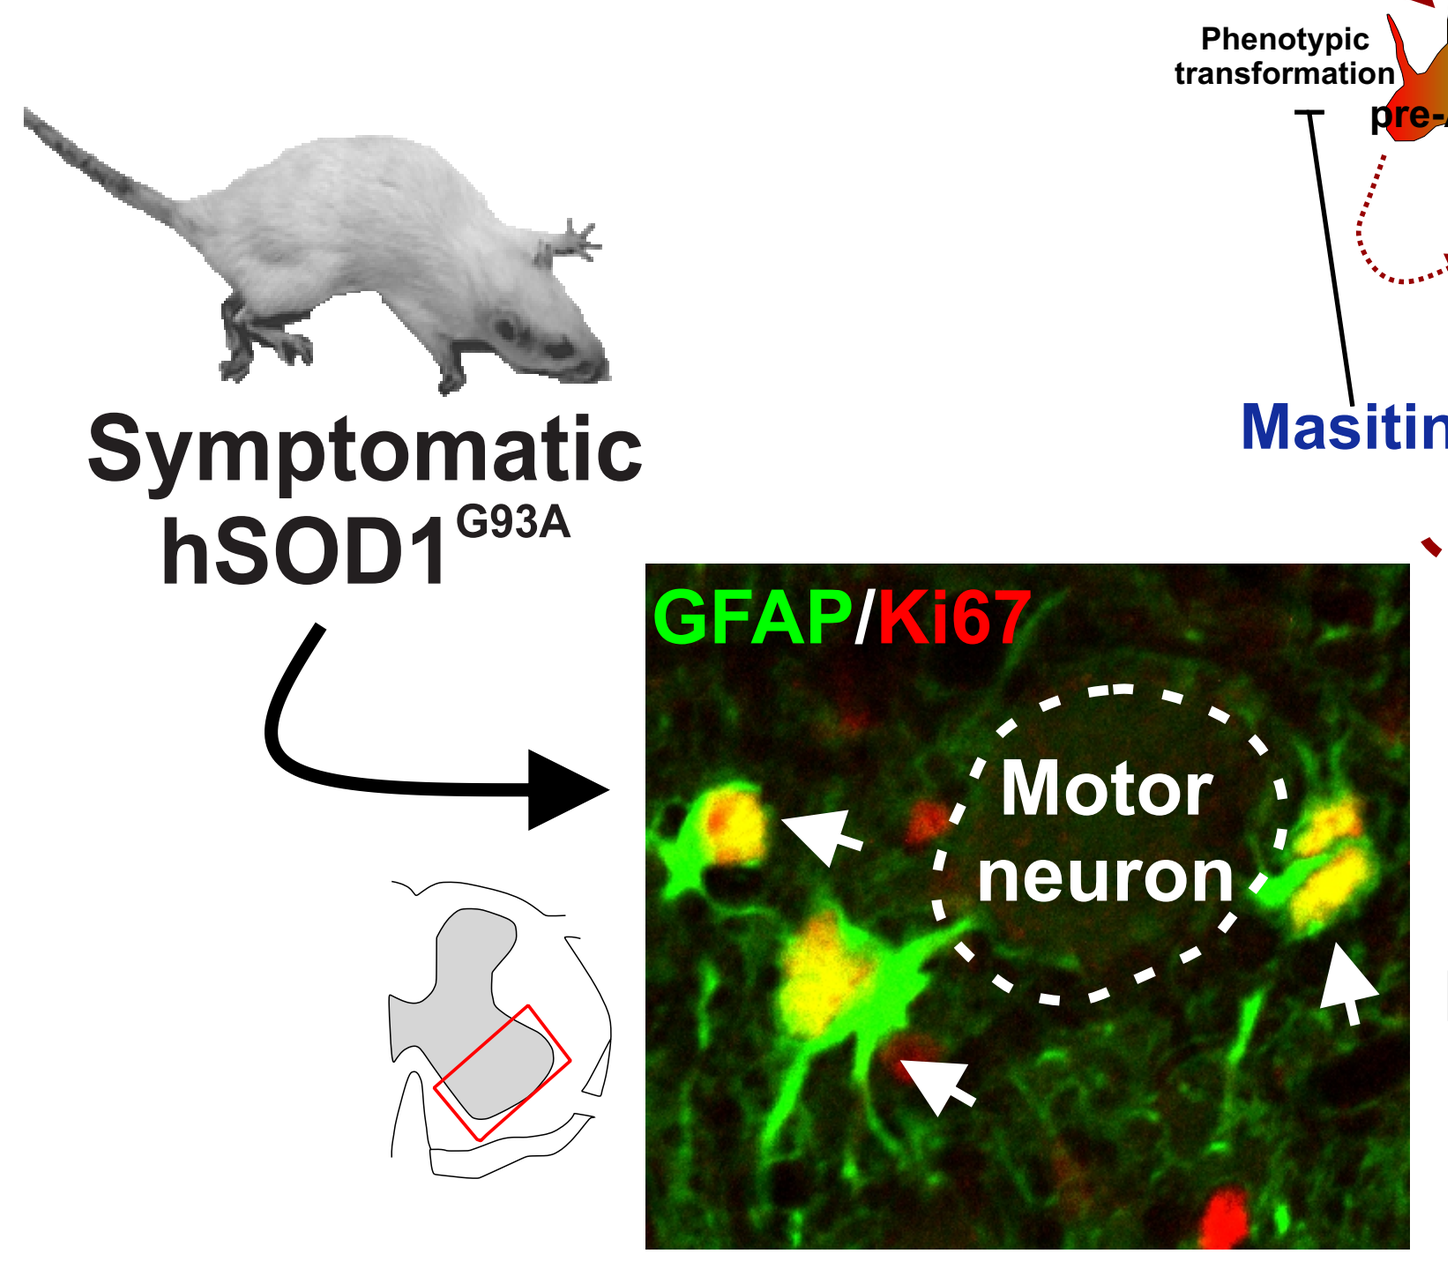
Symptomatic hSOD1G93A
GFAP/Ki67
Motor neuron

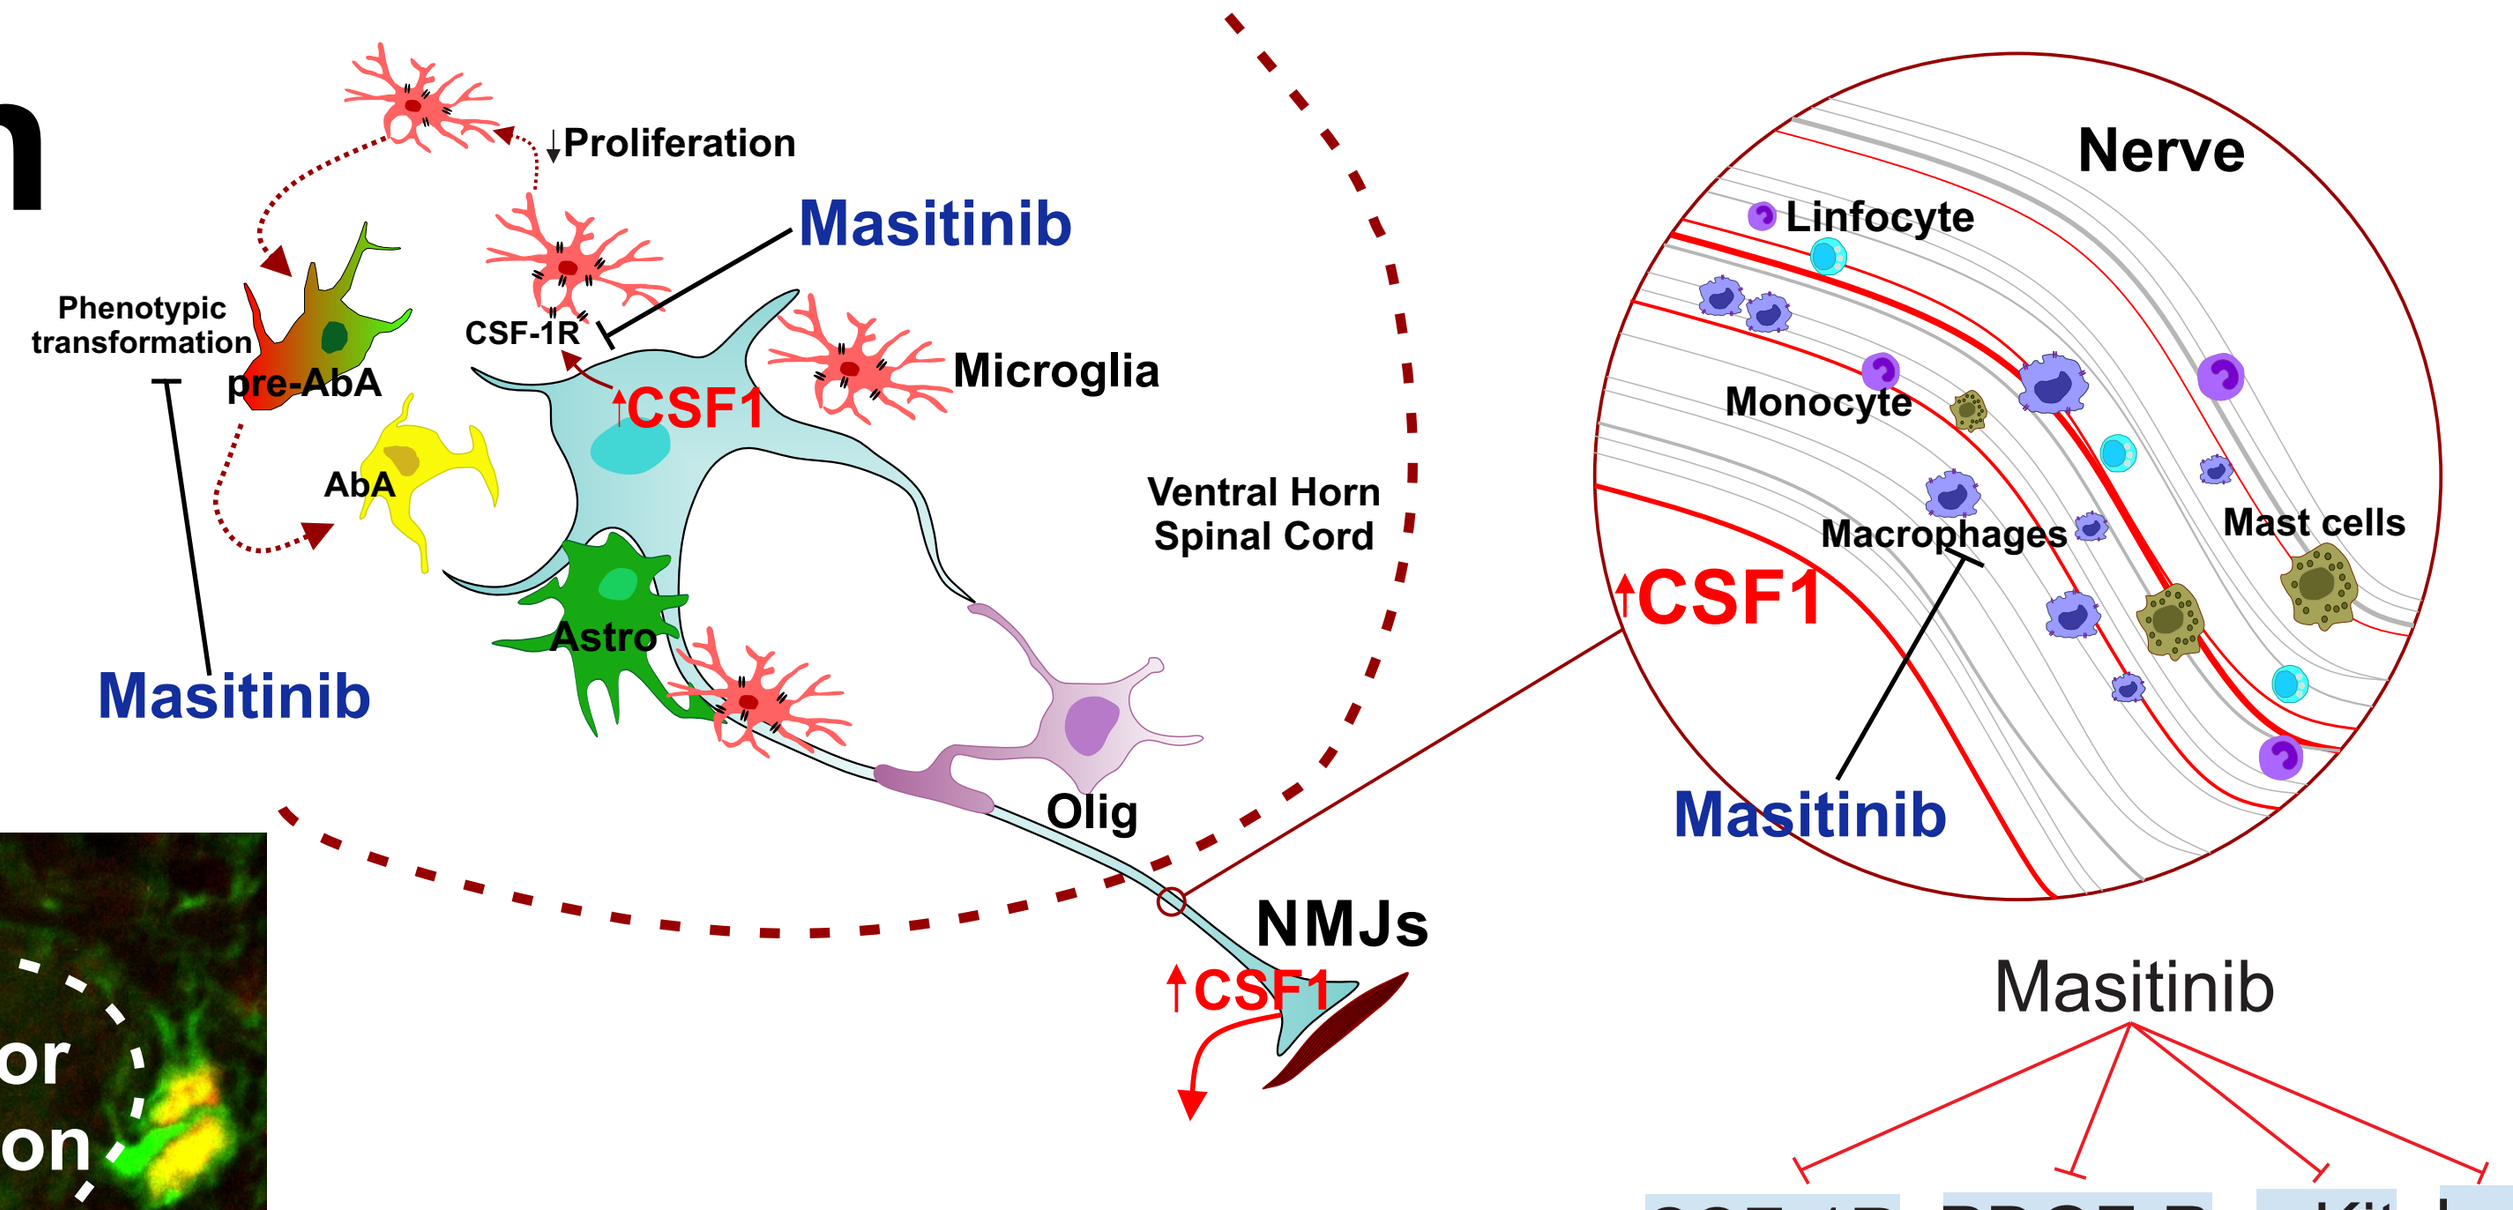
Proliferation
Masitinib
Phenotypic transformation
pre-AbA
CSF-1R
CSF1
Microglia
AbA
Ventral Horn Spinal Cord
Astro
Masitinib
Olig
NMJs
CSF1
Nerve
Linfocyte
Monocyte
Macrophages
Mast cells
CSF1
Masitinib
Masitinib
CSF-1R
PDGF-R
c-Kit
Lyn
Downregulate macrophages/microglia
Downregulate mast cells

Proliferating "aberrant glial cells" in the degenerating spinal cord that induce neurotoxicity

Hypothesis. **Tyrosine kinase inhibition with masitinib might prevent the appearance of aberrant glial cells, regulate inflammation in both central and peripheral nervous systems, and slow paralysis progression.**

Why masitinib?

AB1010 is highly selective kinase inhibitor shown to prevent neuroinflammation in multiple sclerosis, stroke and Alzheimer's disease. Is presently being evaluated in clinical trials for oncologic and neurologic diseases.

Why post-paralysis?

Masitinib targets aberrant glial cells that emerge only after paralysis onset in the SOD1G93A rat model, which is characterized rapidly progressing disease progression.

Results

1 Masitinib inhibits M-CSF induced aberrant glial cell proliferation and inflammatory phenotype

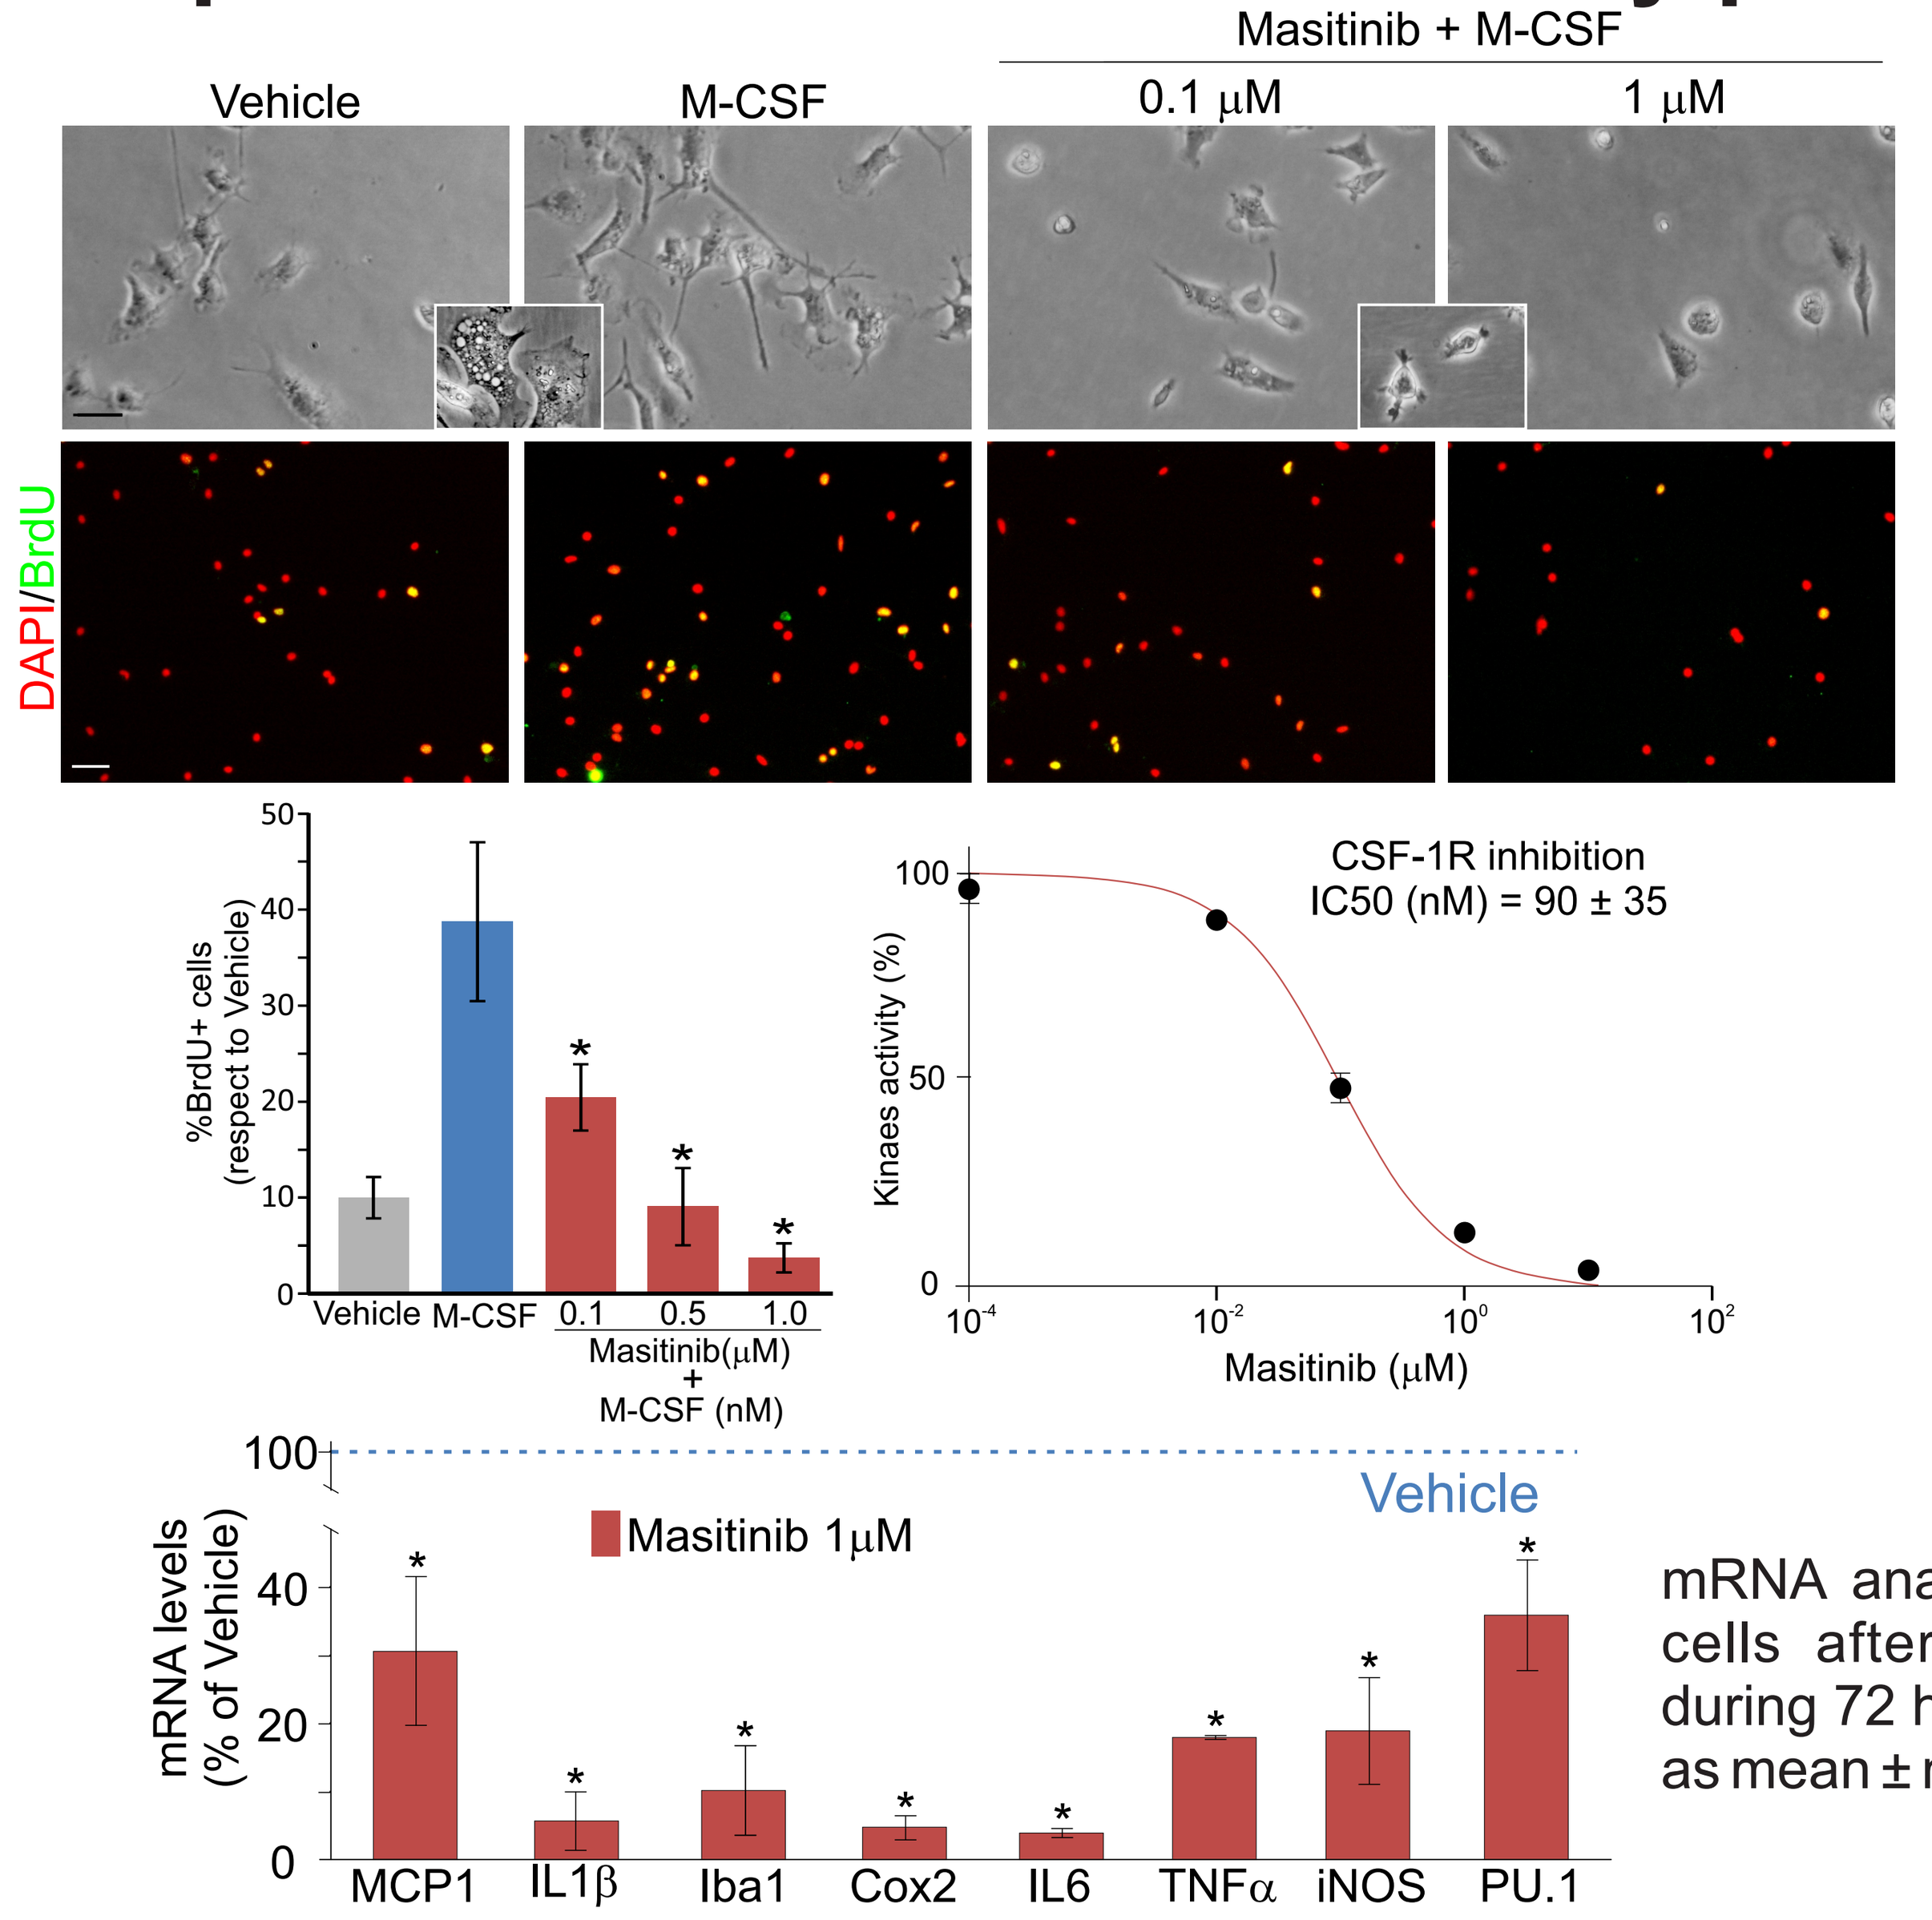
Vehicle
M-CSF
Masitinib + M-CSF
0.1 μM
1 μM
DAPI/BrdU
%BrdU+ cells (respect to Vehicle)
Vehicle M-CSF 0.1 0.5 1.0
Masitinib(μM) + M-CSF (nM)
CSF-1R inhibition
IC50 (nM) = 90 ± 35
Kinaes activity (%)
Masitinib (μM)
Vehicle
Masitinib 1μM
mRNA levels (% of Vehicle)
MCP1
IL1β
Iba1
Cox2
IL6
TNFα
iNOS
PU.1

mRNA analysis of aberrant glial cells after Masitinib treatment during 72 hs. Data are expressed as mean ± m.s.e. *p<0.01.

2 Masitinib reduces aberrant glial cells in the degenerating spinal cord

Methods. Daily masitinib 30 mg/kg vs Vehicle, orally.

Treatment starting day 1 post-paralysis and lasting 20 days.

Hindlimb onset only

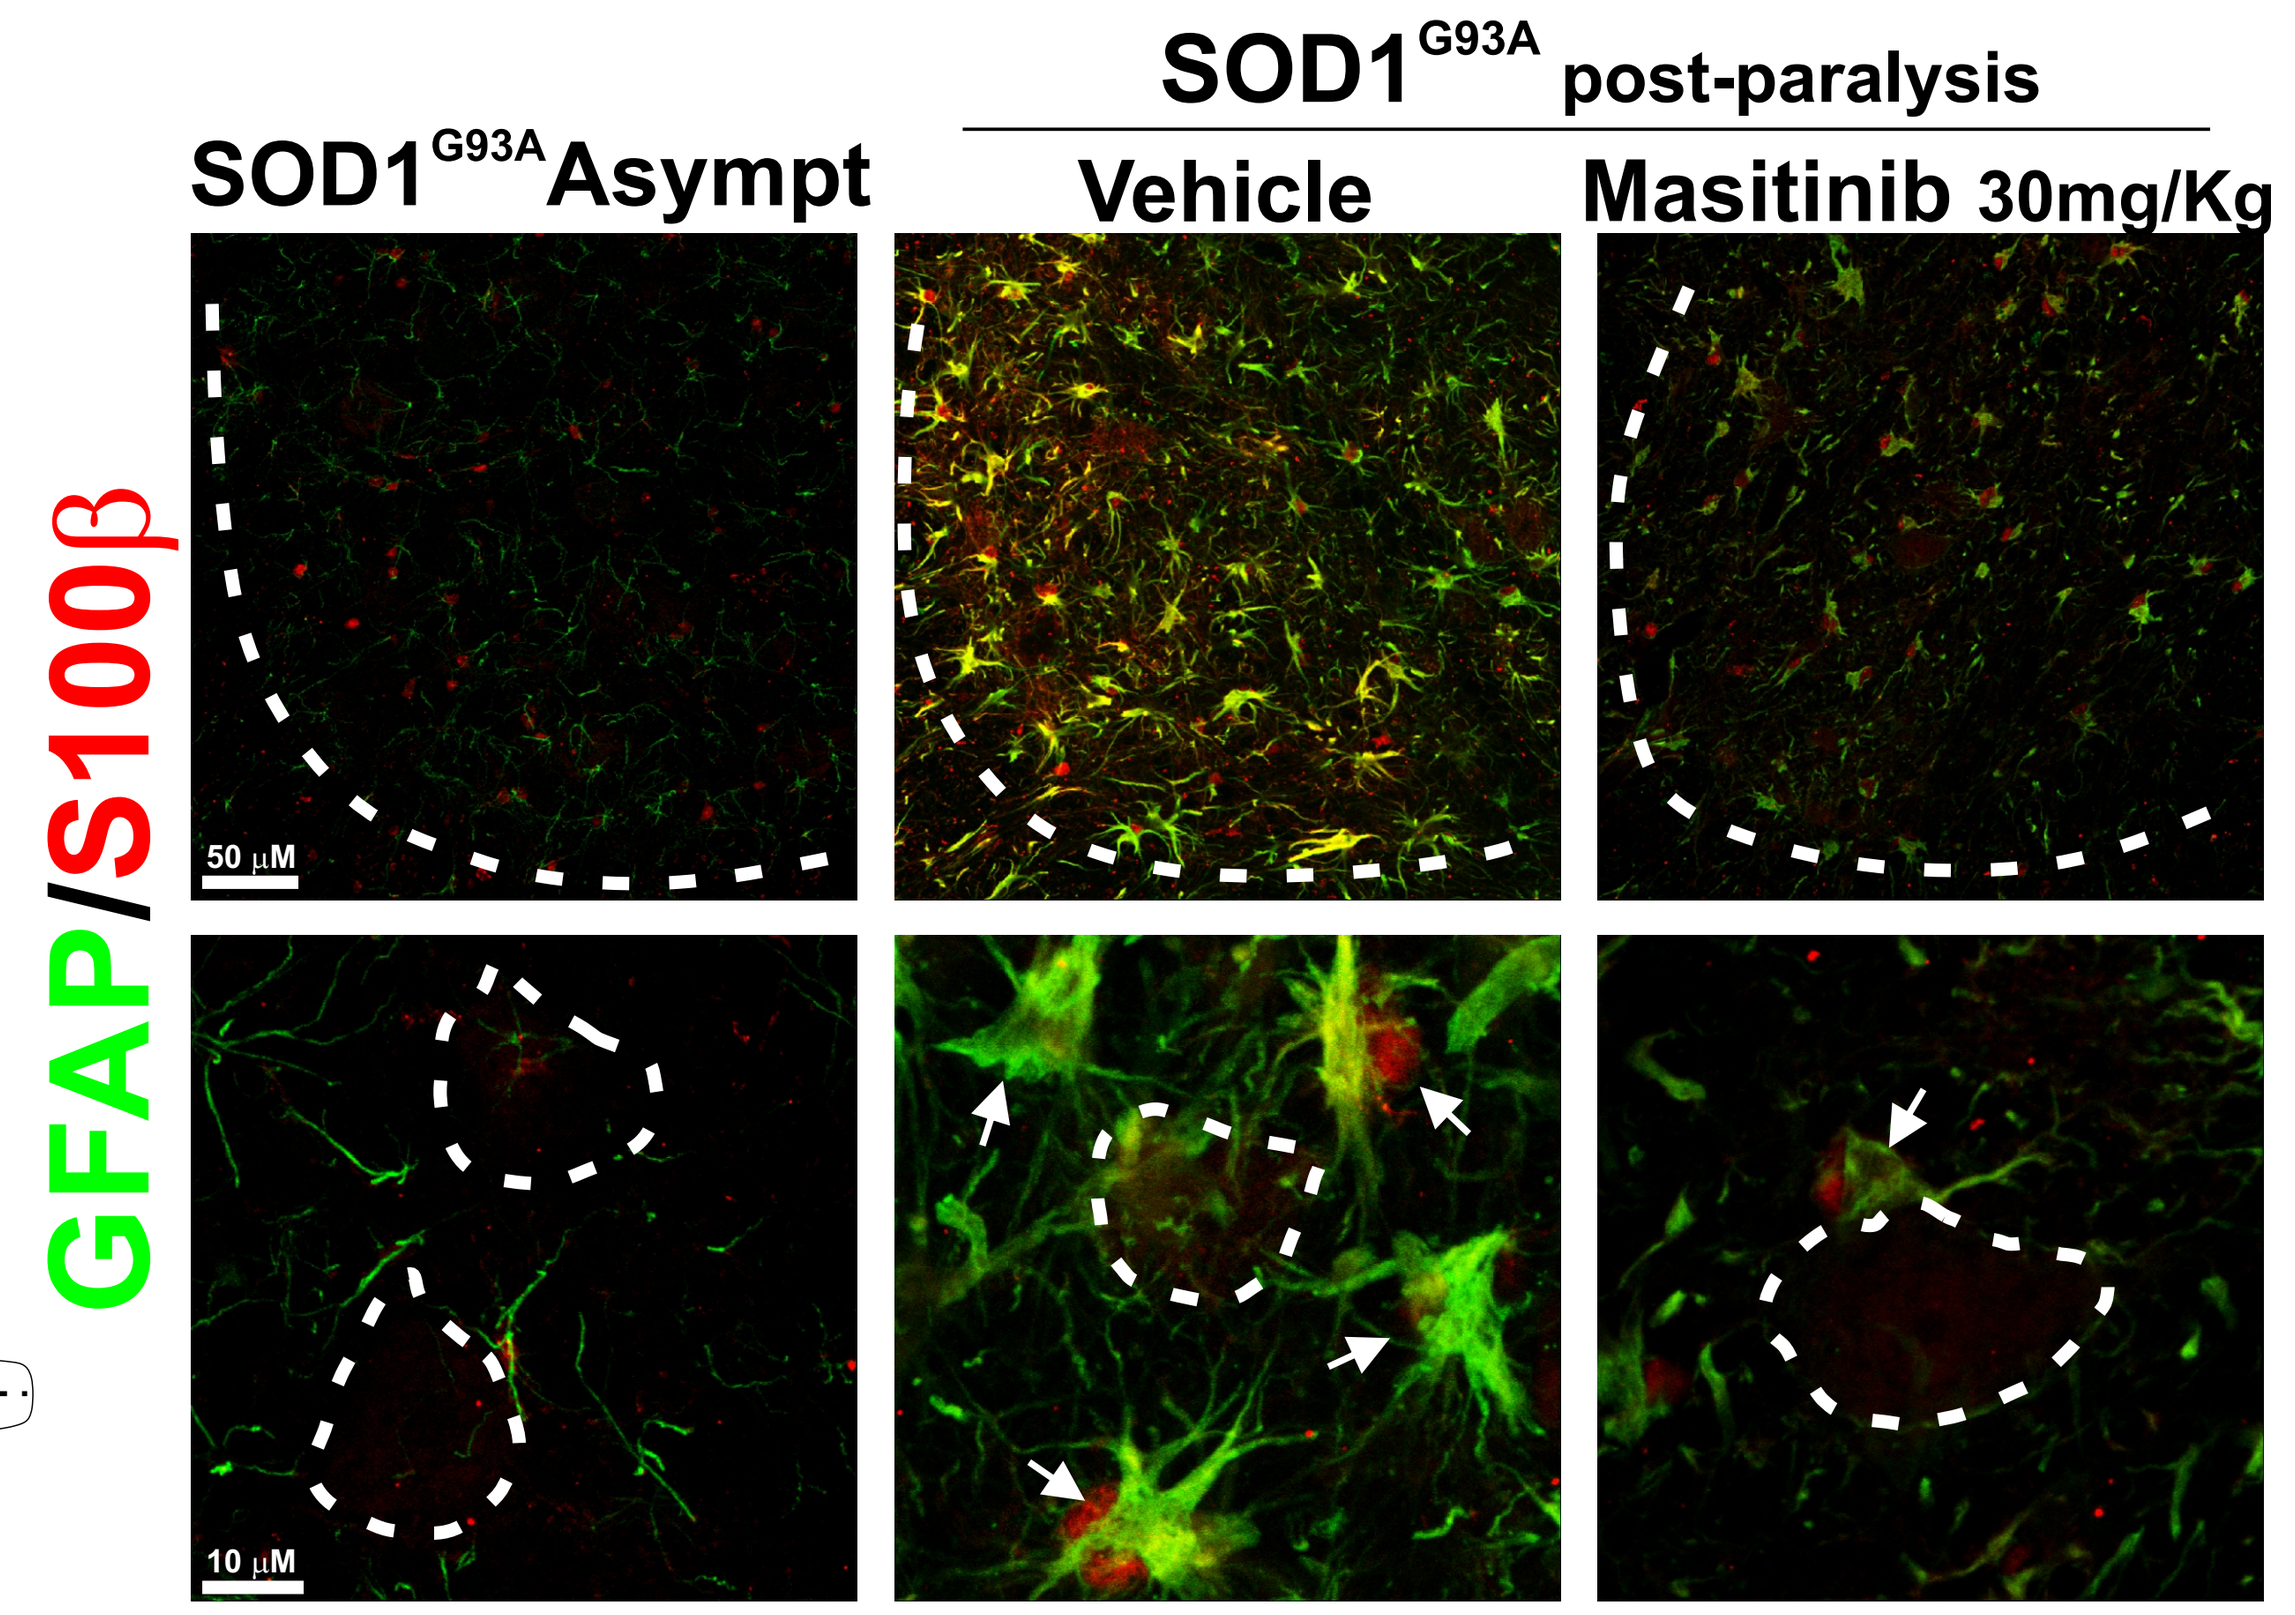
SOD1G93A Asympt
SOD1G93A post-paralysis
Vehicle
Masitinib 30mg/Kg
GFAP/S100β
50 μM
10 μM

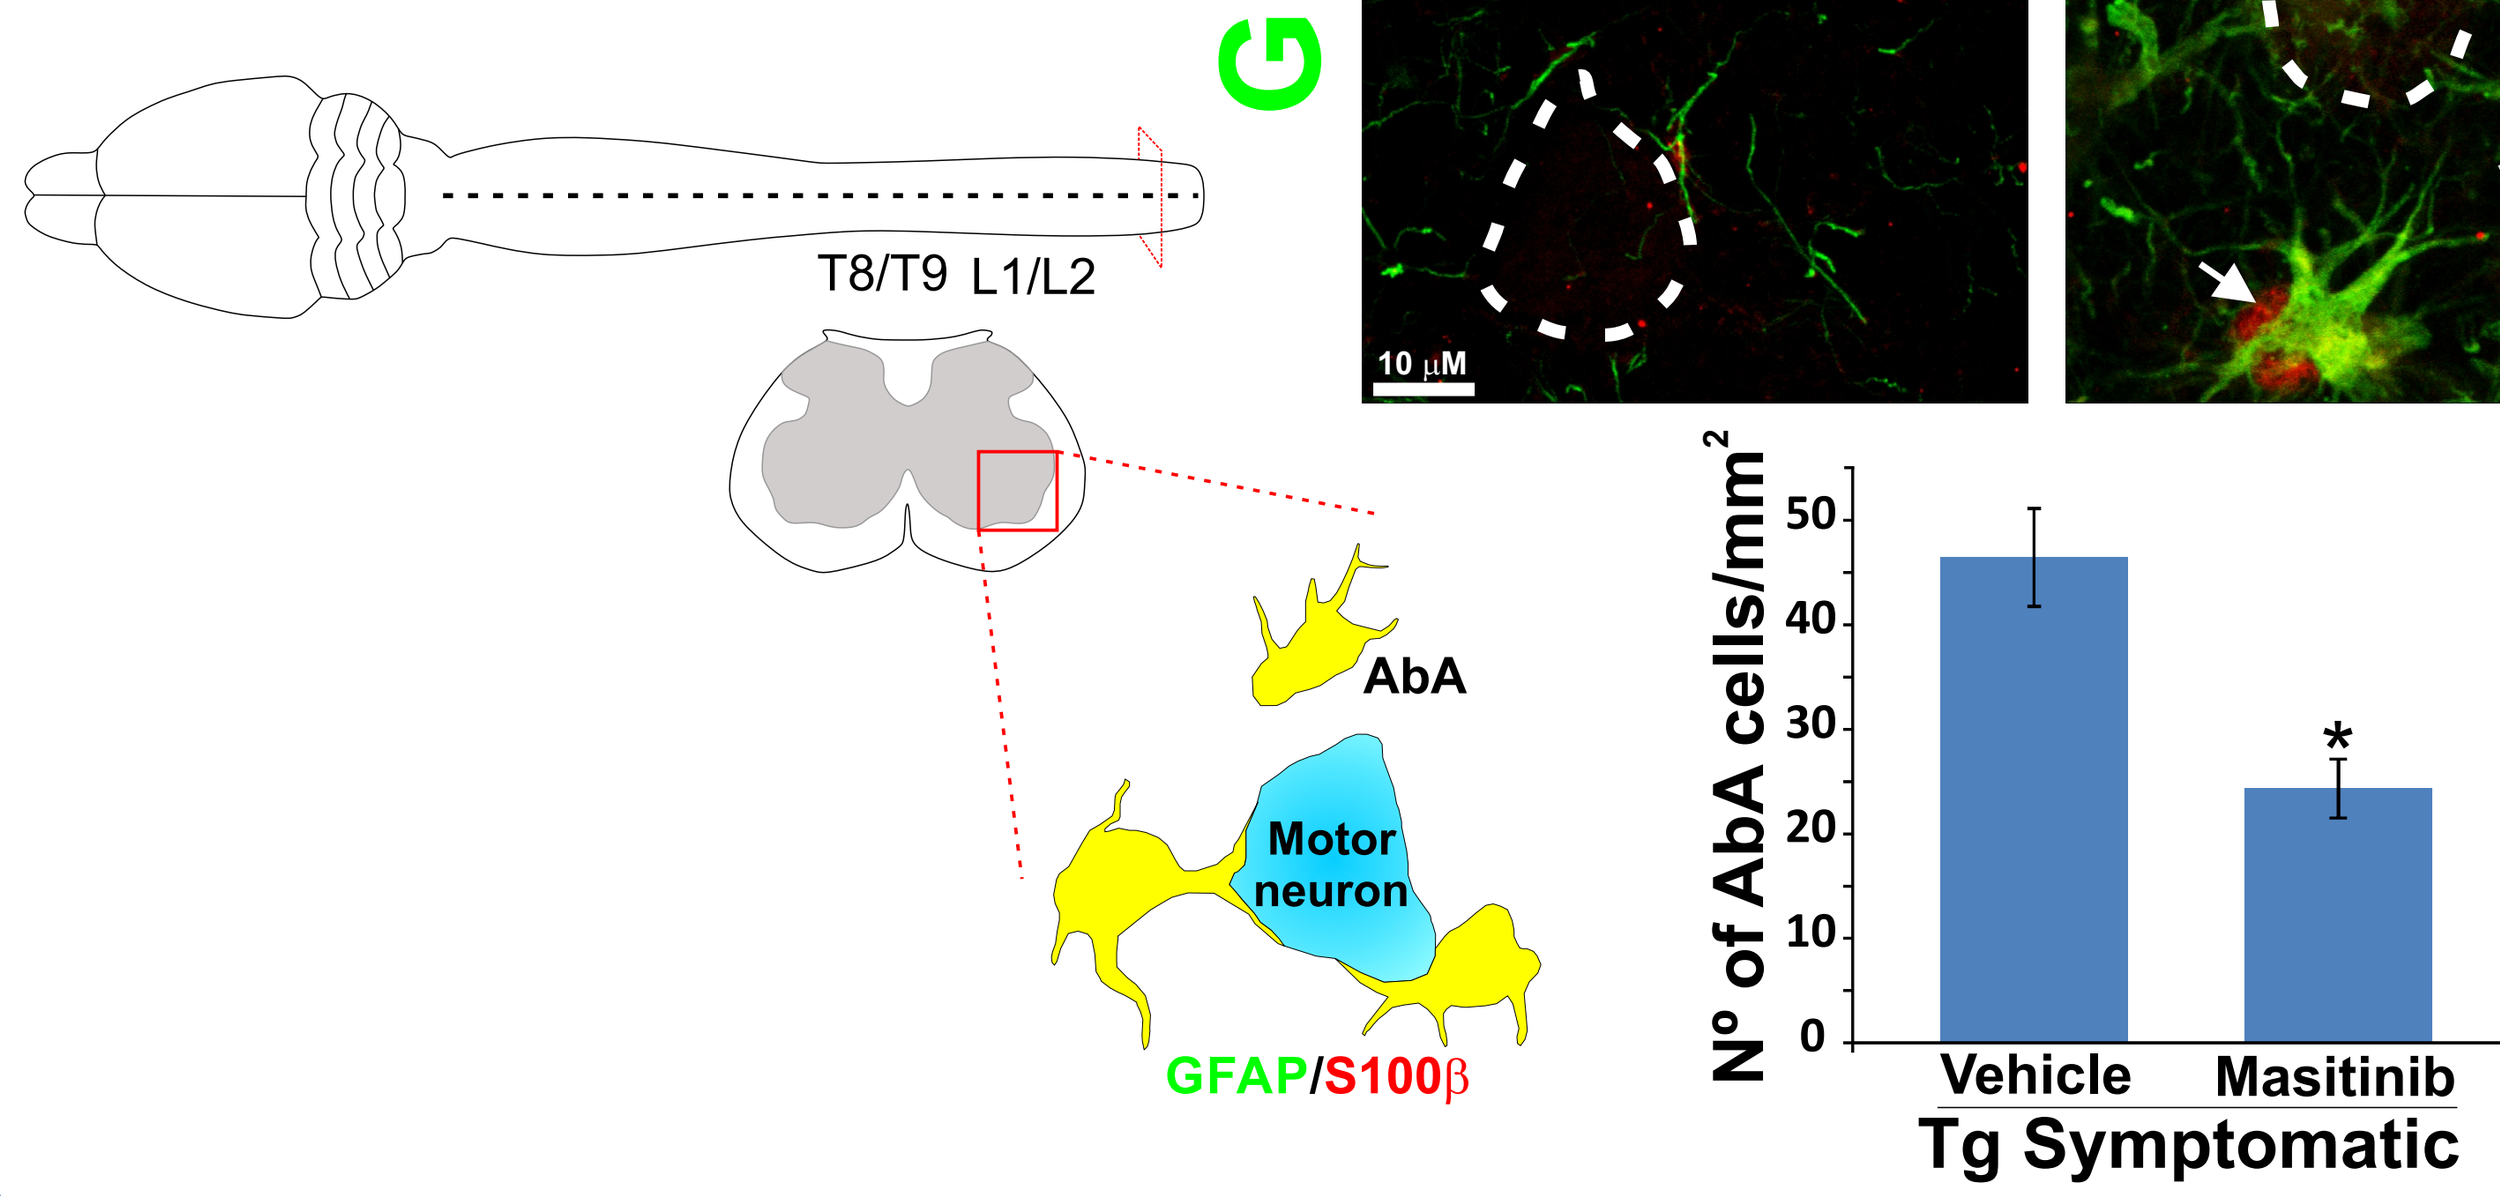
T8/T9 L1/L2
AbA
Motor neuron
GFAP/S100β
Nº of AbA cells/mm2
Vehicle
Masitinib
Tg Symptomatic

3 Masitinib prevents microglia activation and motor neuron pathology in ALS rats

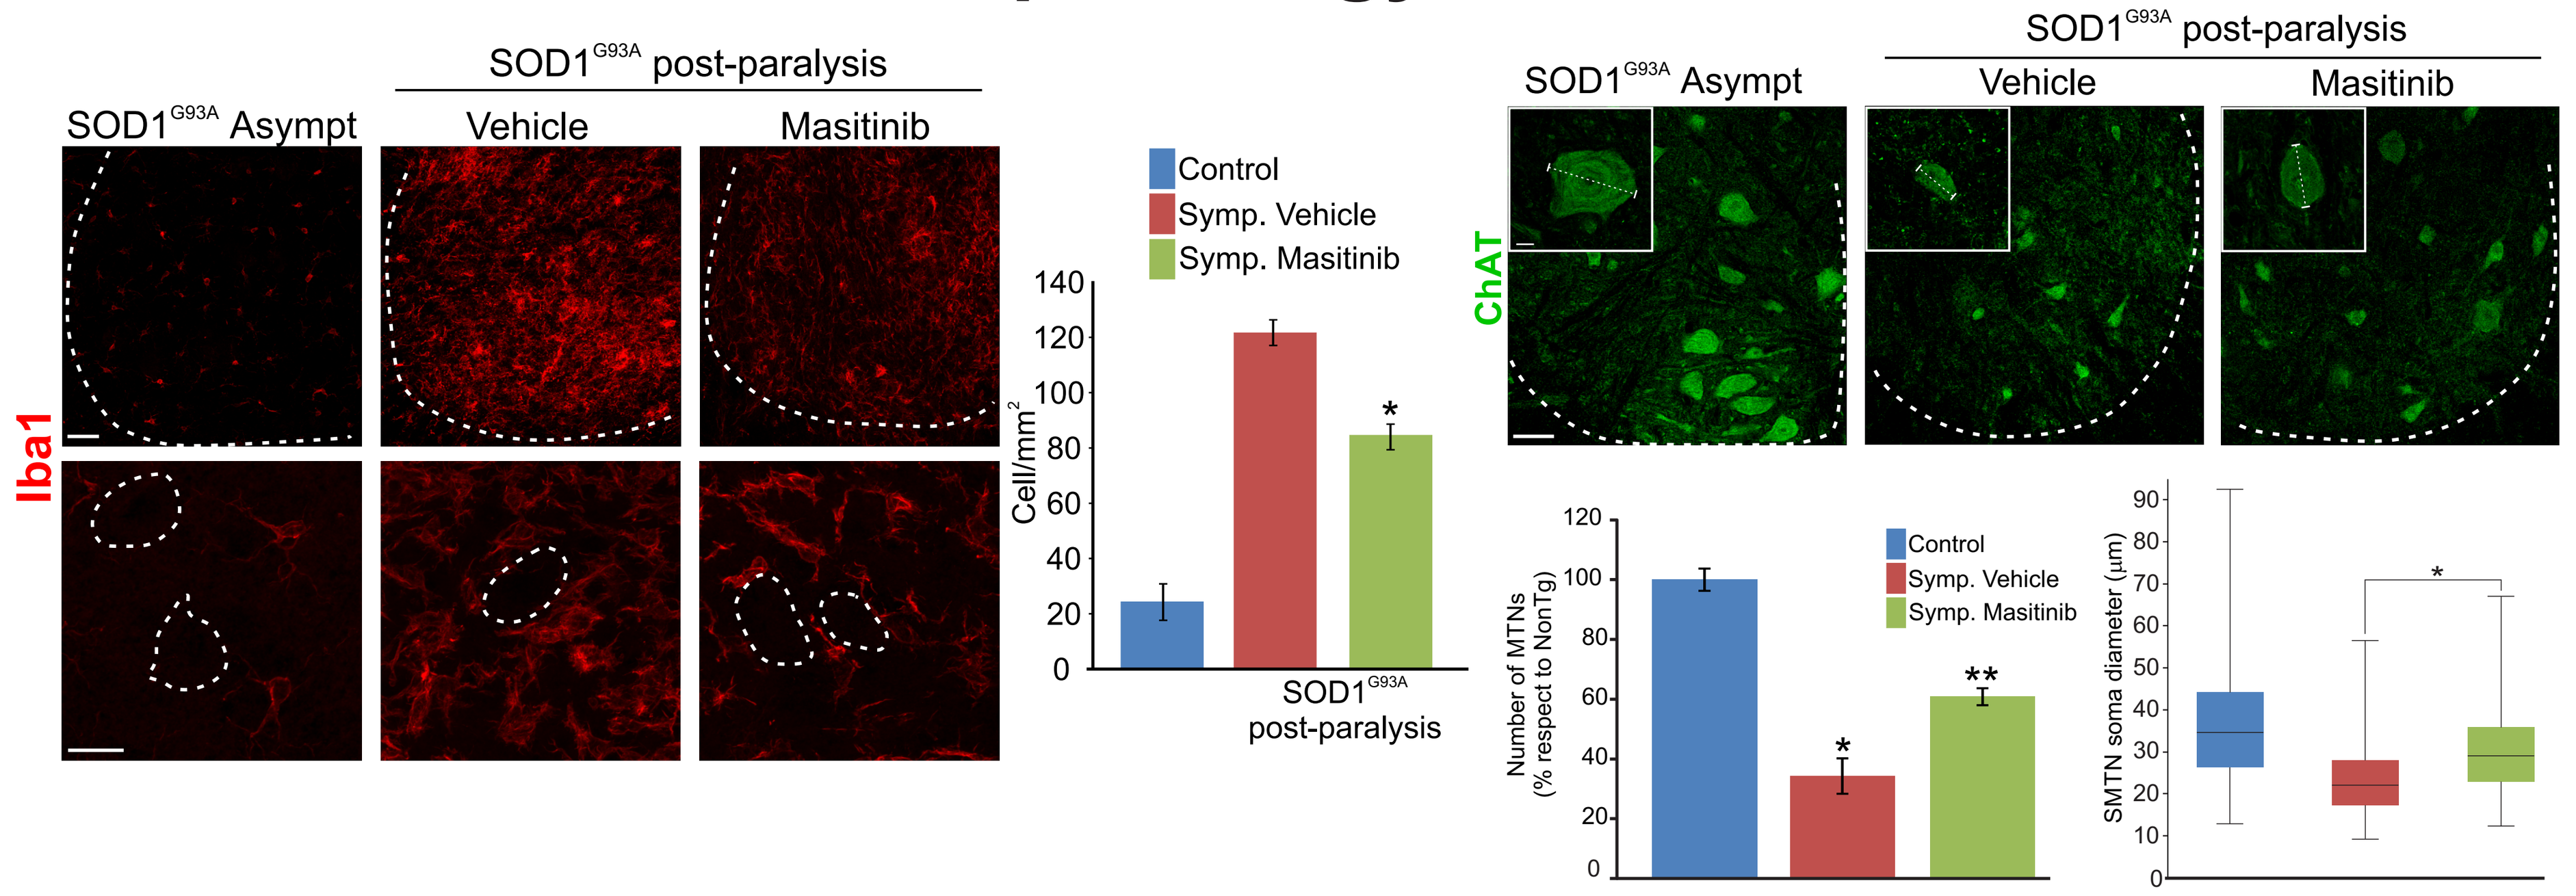
SOD1G93A Asympt
SOD1G93A post-paralysis
Vehicle
Masitinib
Iba1
Control
Symp. Vehicle
Symp. Masitinib
Cell/mm2
SOD1G93A post-paralysis
ChAT
Number of MTNs (% respect to NonTg)
SMTN soma diameter (μm)

4 M-CSF expression in the degenerating sciatic nerve

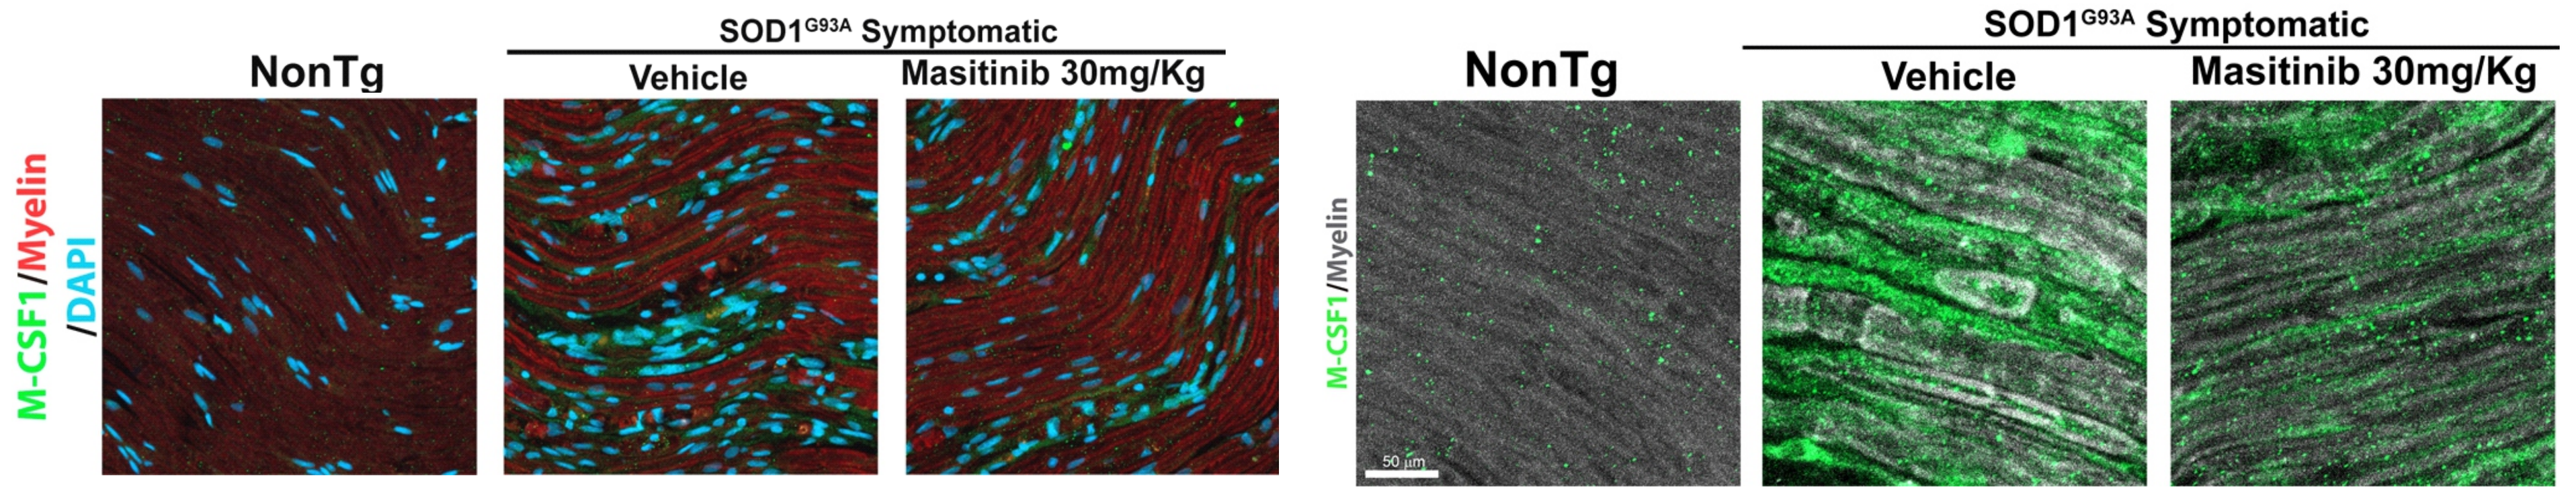
NonTg
SOD1G93A Symptomatic
Vehicle
Masitinib 30mg/Kg
M-CSF1/Myelin/DAPI
M-CSF1/Myelin
50 μm

5 Masitinib reduces macrophages and mast cell cell inflitration in sciatic nerve

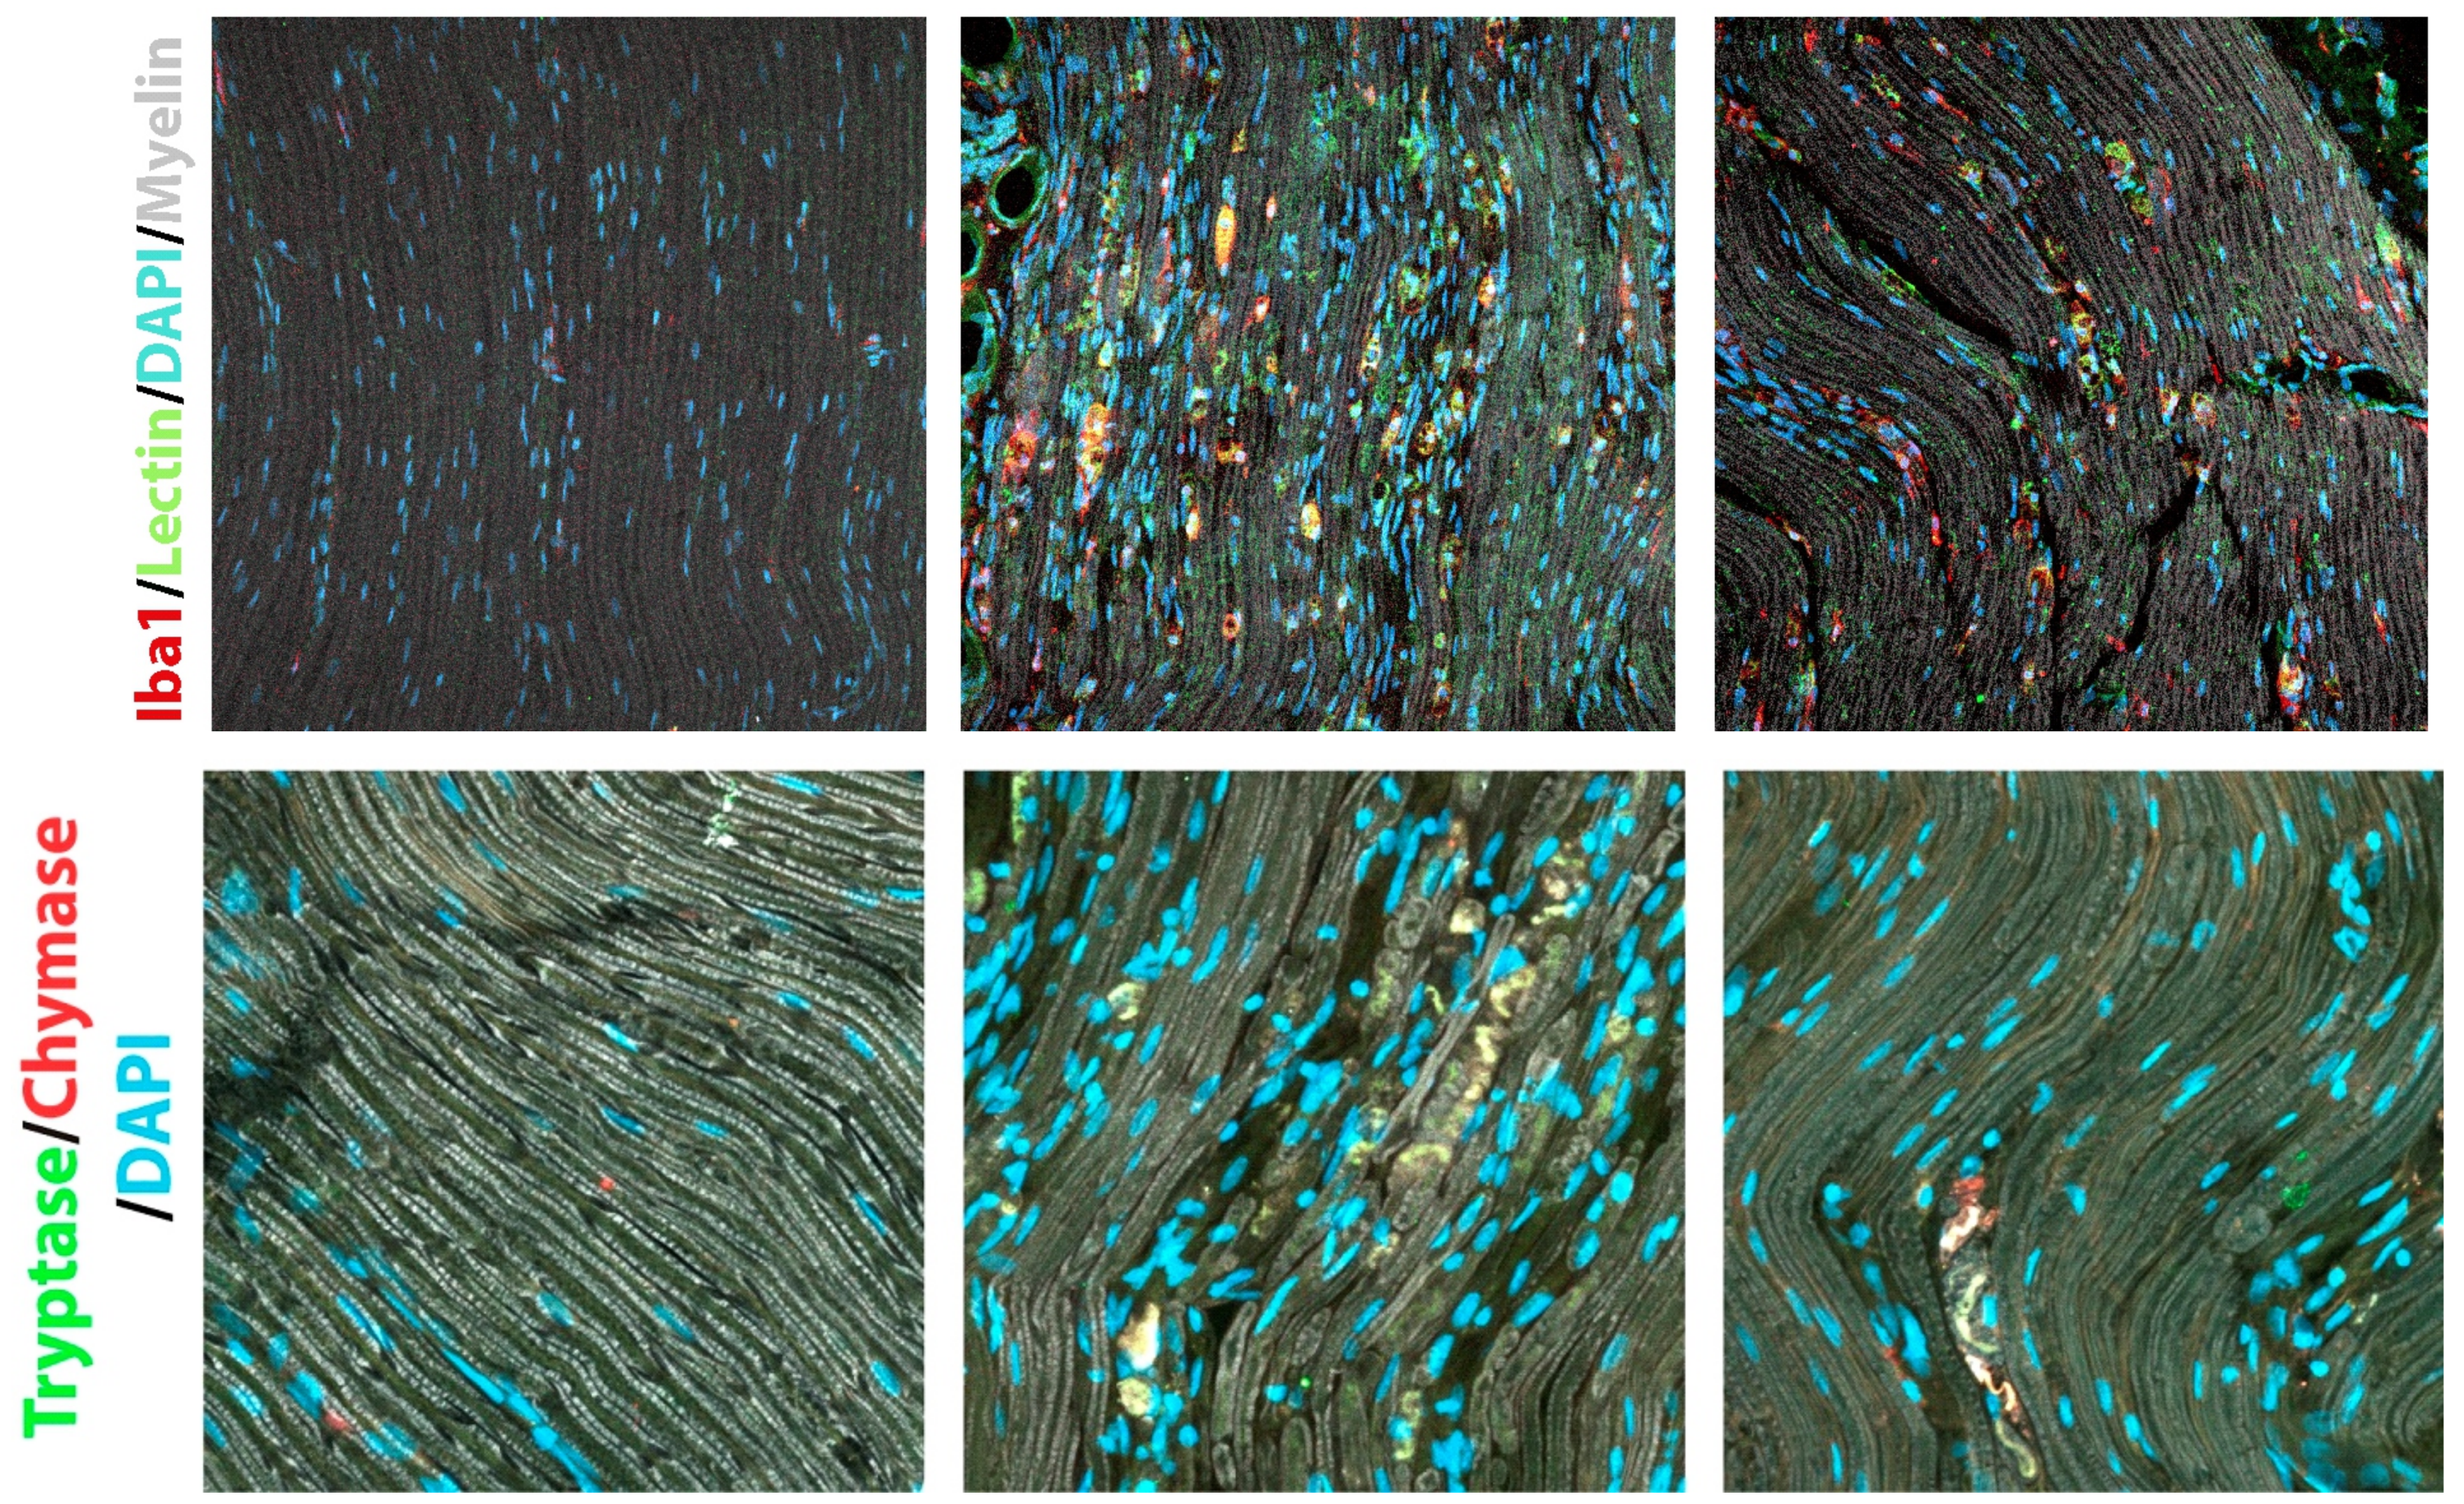
Iba1/Lectin/DAPI/Myelin
Tryptase/Chymase/DAPI

6 Masitinib increases post-paralysis survival

Methods. Daily oral masitinib (30mg/kg) or vehicle administration, starting after paralysis onset.

Survival was increased even when treatment was started 7 days after paralysis onset (7d onset).

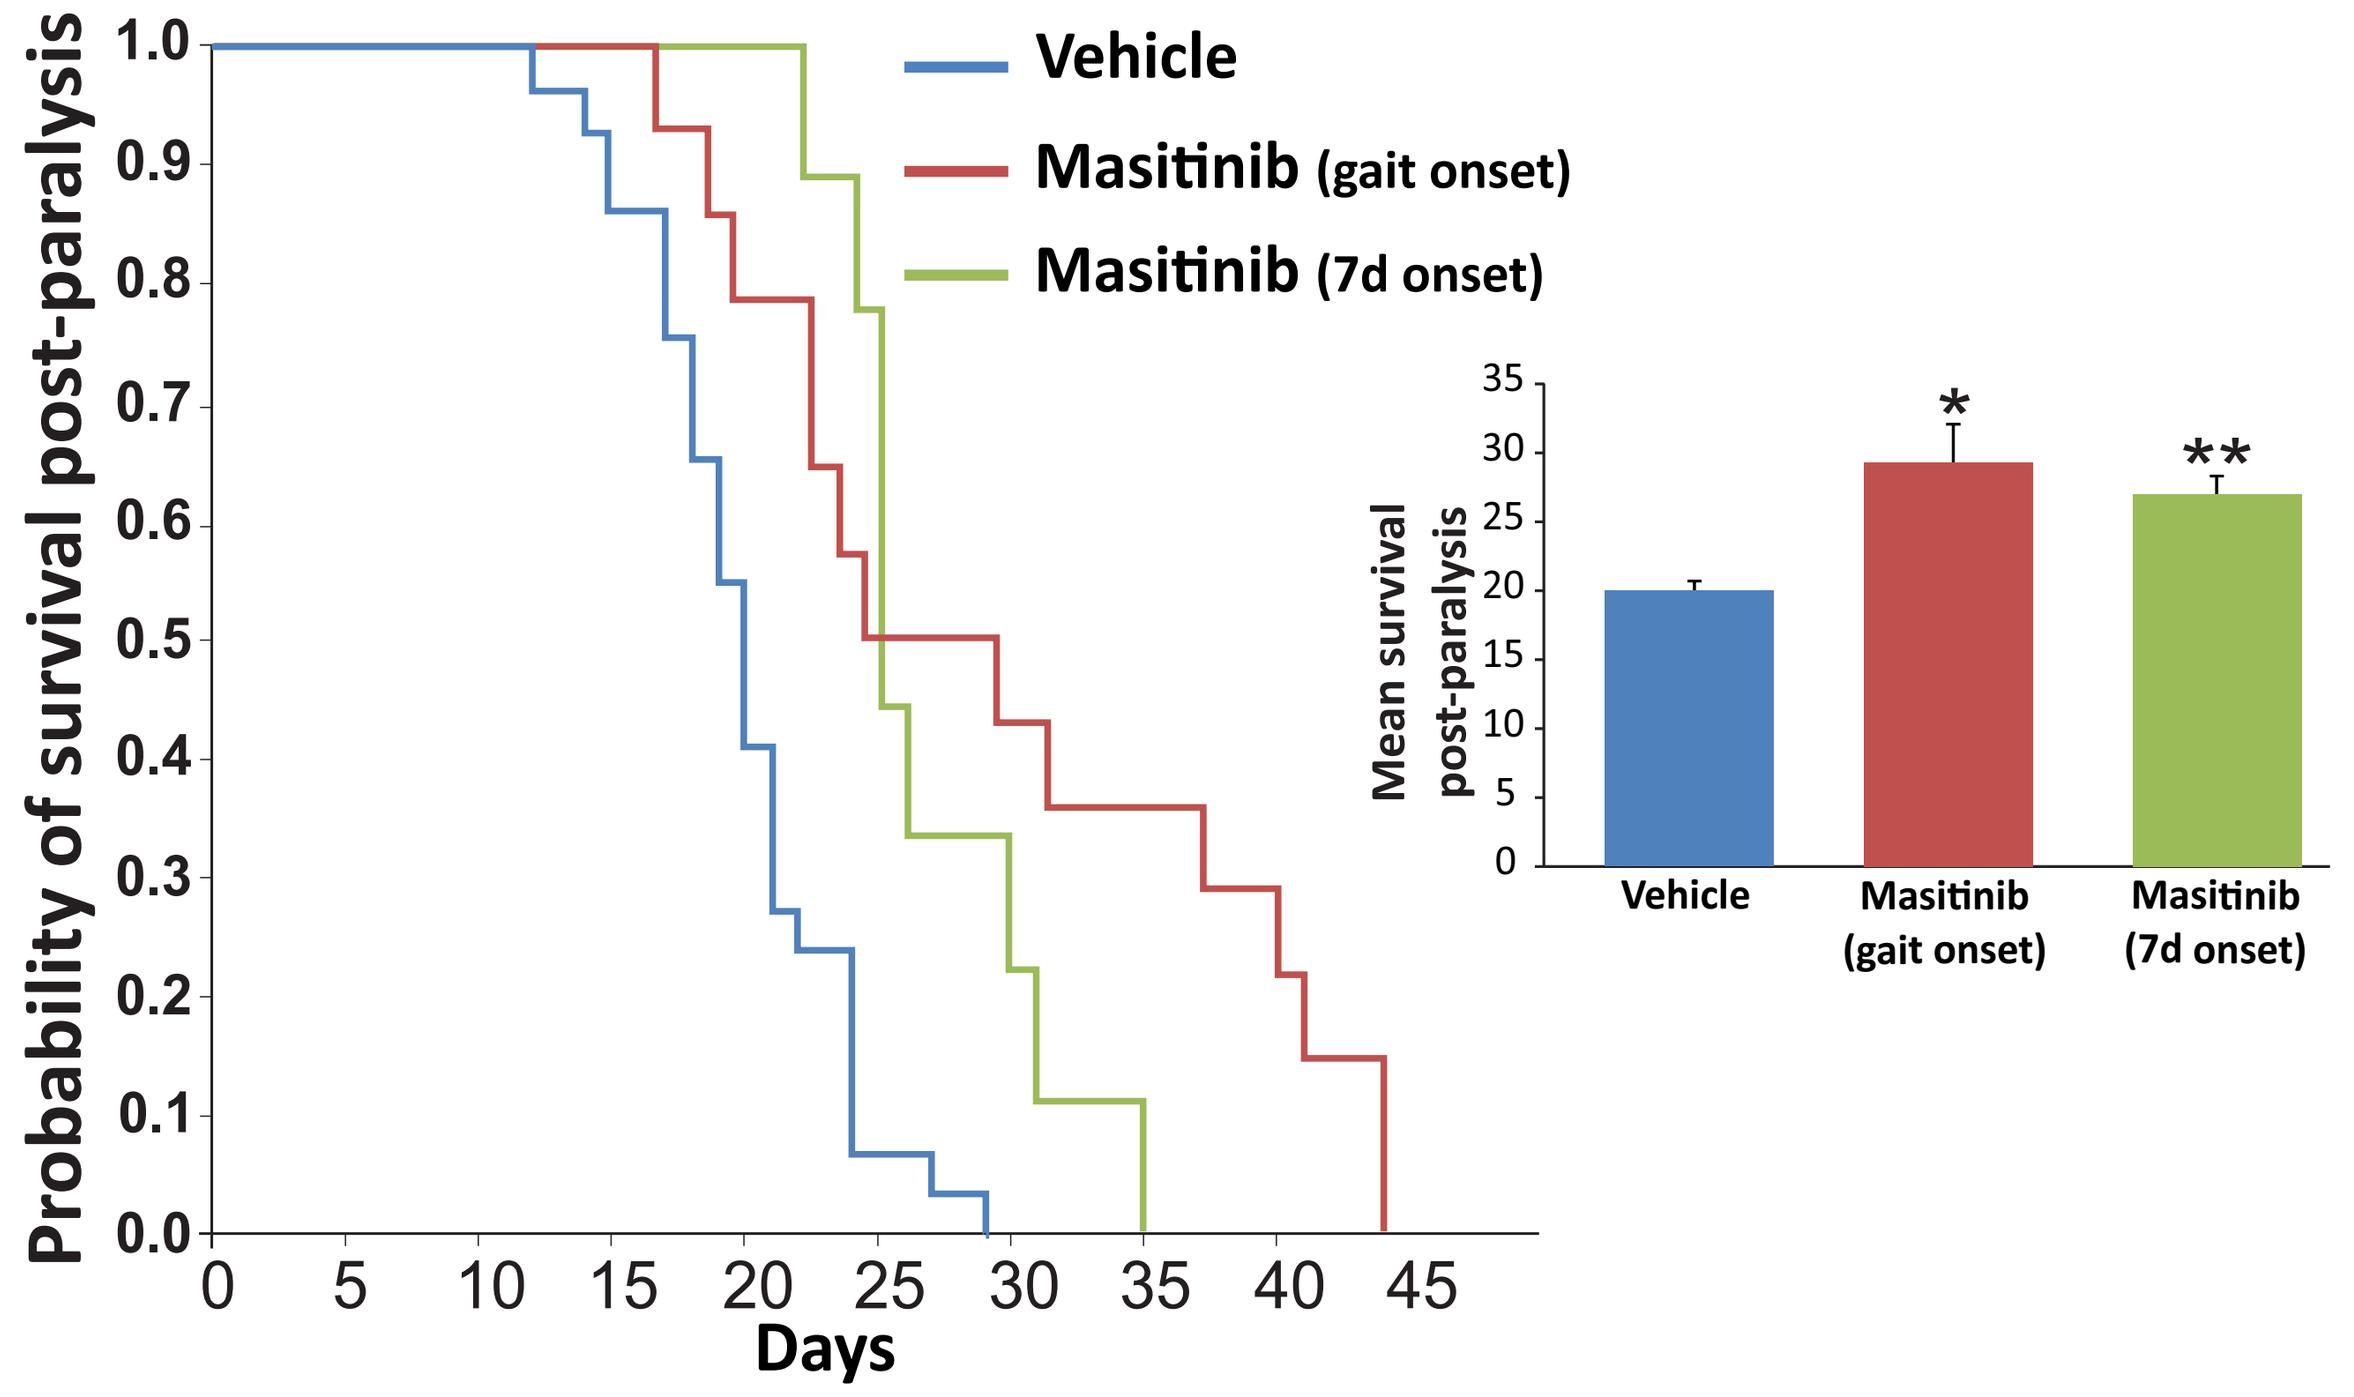
Probability of survival post-paralysis
Days
Vehicle
Masitinib (gait onset)
Masitinib (7d onset)
Mean survival post-paralysis
Vehicle
Masitinib (gait onset)
Masitinib (7d onset)

Conclusions

- Masitinib **can slow paralysis progression** by targeting CNS aberrant glial cells and inflammatory cell infiltration in the PNS.
- Protective effect would be at least in part mediated by **masitinib inhibition of CSF-1R (microglia/macrophages) and c-Kit (mast cells)**
- Remarkably the protection can be obtained **when masitinib is administered post-paralysis.**
- Masitinib represents a new drug candidate for the treatment of ALS.

References

- Díaz-Amarilla P, Olivera-Bravo S, Trias E, Cragnolini A, Martínez-Palma L, Cassina P, Beckman J, Barbeito L. Phenotypically aberrant astrocytes that promote motoneuron damage in a model of inherited amyotrophic lateral sclerosis. Proc Natl Acad Sci USA. 2011 Nov 1;108(44):18126-31.
- Trias E, Díaz-Amarilla P, Olivera-Bravo S, Isasi E, Drechsel DA, Lopez N, Bradford CS, Ireton KE, Beckman JS, Barbeito L. Phenotypic transition of microglia into astrocyte-like cells associated with disease onset in a model of inherited ALS. Front Cell Neurosci. 2013 Dec 24;7:274.